Curriculum-Based Online Education to Improve Provider Knowledge, Competence, and Confidence in the Current and Future Treatment Landscape for Patients Living With Schizophrenia

Wendy Warfield, MHA, OTR/L; Lisette T. Arnaud-Hevi, PhD; Jovanna Lubarda, PhD: Medscape LLC, Newark, NJ, USA

Presented at **Psych Congress 2024,** October 29-November 1, 2024; Poster #190

BACKGROUND

Purpose: To examine whether a continuing medical education (CME) curriculum intervention can improve provider knowledge, competence, and confidence in individualizing treatment plans for patients living with Schizophrenia

METHODS

Providers were exposed to an educational curriculum consisting of 6 CME-certified activities offered in multiple delivery formats. Data for psychiatrists and primary care providers were collected for ≤ 12 weeks following each activity. A 3-item, multiple choice, knowledge/competence questionnaire and 1 confidence assessment question were administered before and after each activity. Physicians who completed questions both pre- and post-assessment were aggregated across activities, stratified by learning theme. McNemar's test was used to assess statistical significance ($P < .05$).

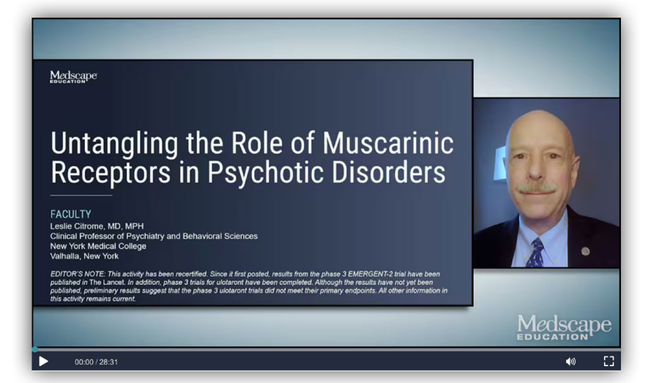
Untangling the Role of Muscarinic Receptors in Psychotic Disorders

Untangling the Role of Muscarinic Receptors in Psychotic Disorders

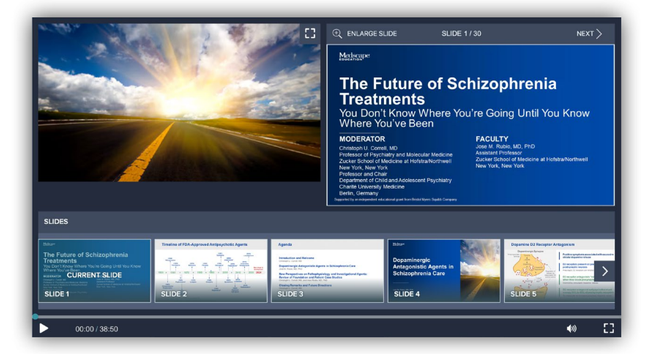
The Future of Schizophrenia Treatments

The Future of Schizophrenia Treatments

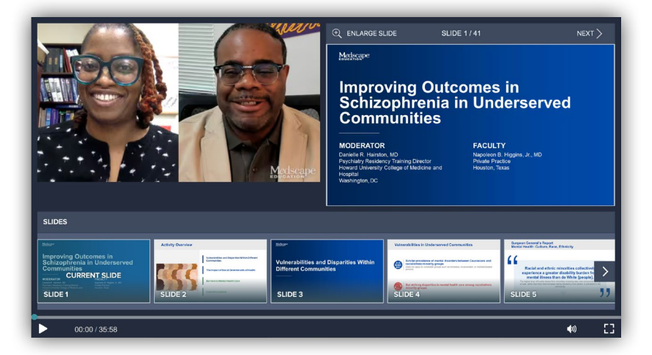
Improving Outcomes in Schizophrenia in Underserved Communities

Improving Outcomes in Schizophrenia in Underserved Communities

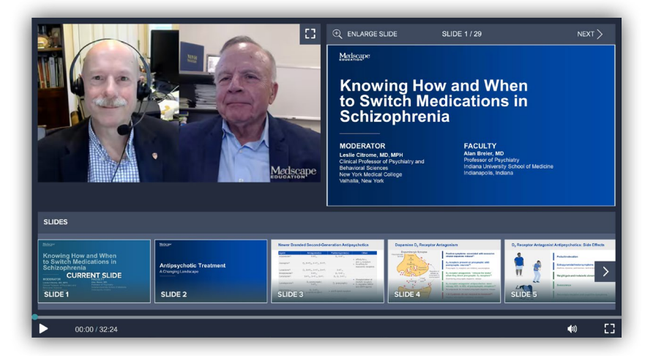
Knowing How and When to Switch Medications in Schizophrenia

Knowing How and When to Switch Medications in Schizophrenia

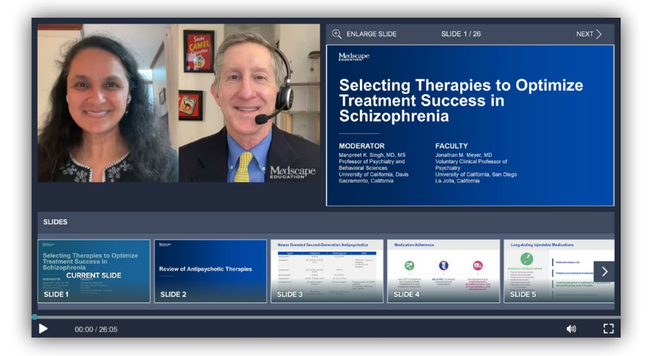
Selecting Therapies to Optimize Treatment Success in Schizophrenia

Selecting Therapies to Optimize Treatment Success in Schizophrenia

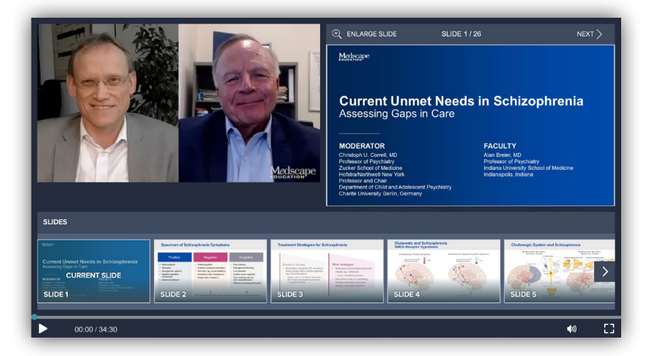
Current Unmet Needs in Schizophrenia

Current Unmet Needs in Schizophrenia

RESULTS

Participation in a CME-certified curriculum significantly improved knowledge/competence and confidence among psychiatrist and primary care providers related to individualized care for their patients with schizophrenia.

Knowledge, Competence & Confidence, by Specialty

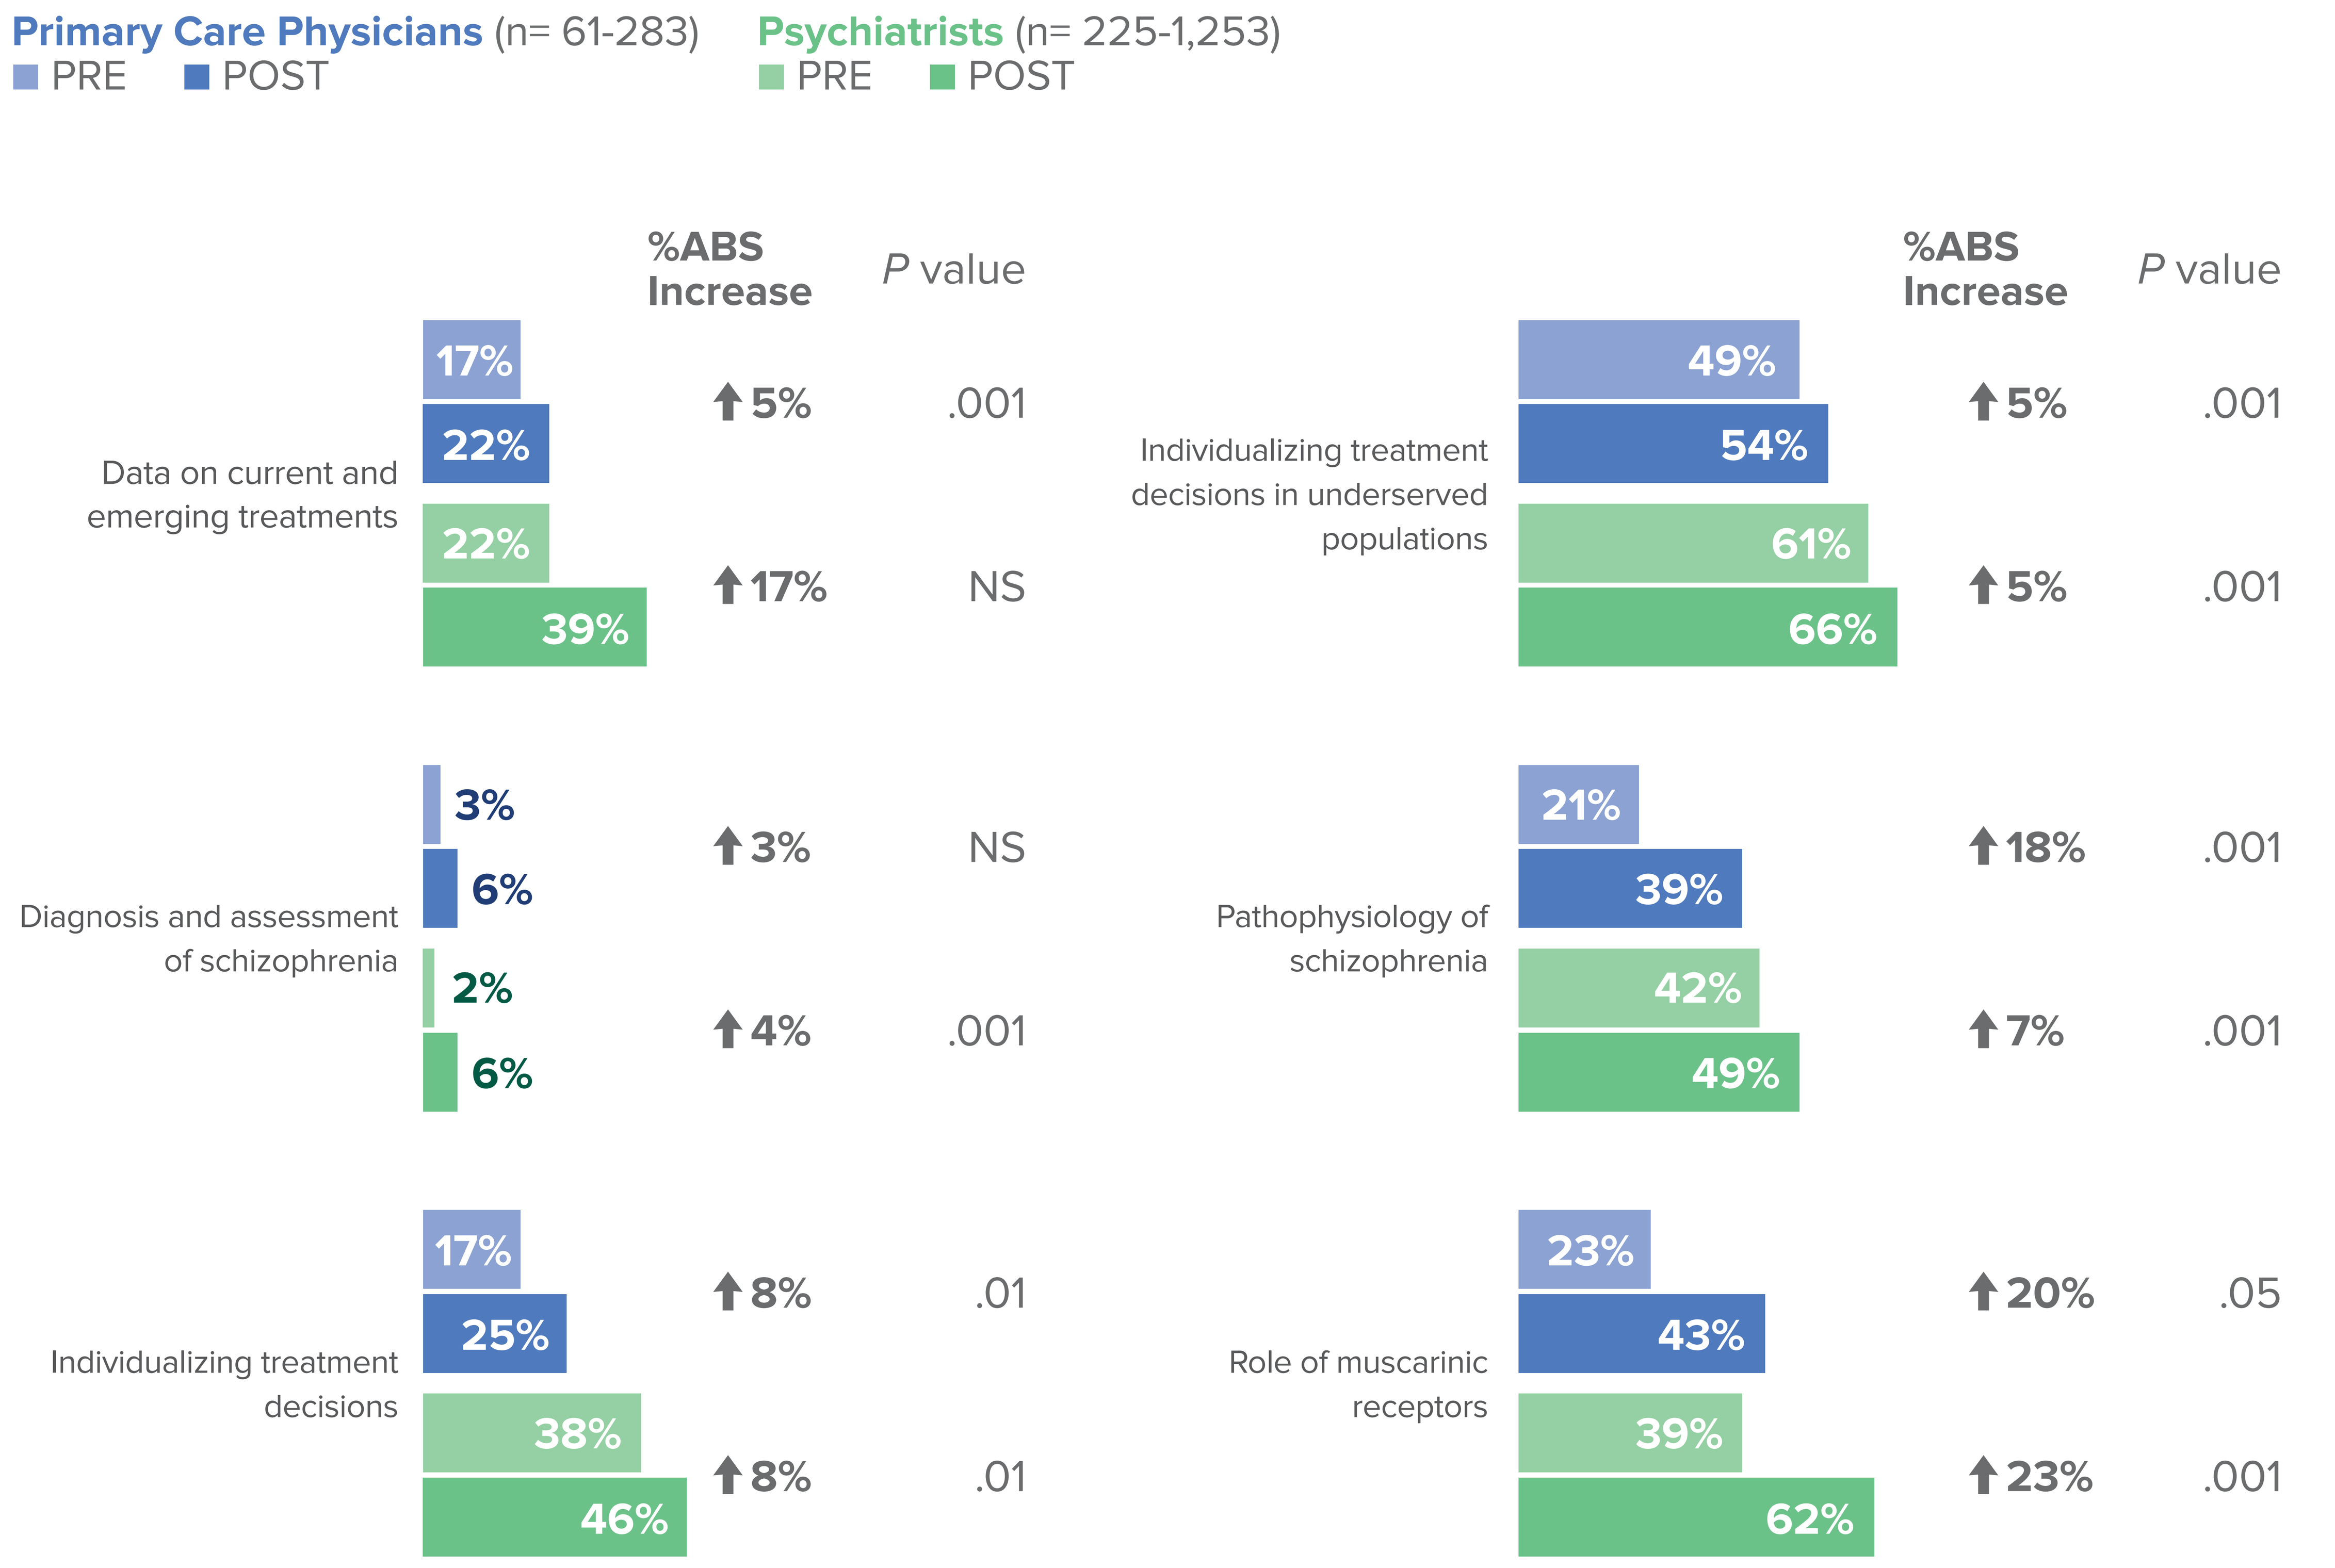
Primary Care Physicians (n= 61-283)
PRE
POST
Psychiatrists (n= 225-1,253)
PRE
POST
%ABS Increase
P value
Data on current and emerging treatments
17%
22%
22%
39%
5%
.001
17%
NS
Diagnosis and assessment of schizophrenia
3%
6%
2%
6%
3%
NS
4%
.001
Individualizing treatment decisions
17%
25%
38%
46%
8%
.01
8%
.01
Individualizing treatment decisions in underserved populations
49%
54%
61%
66%
5%
.001
5%
.001
Pathophysiology of schizophrenia
21%
39%
42%
49%
18%
.001
7%
.001
Role of muscarinic receptors
23%
43%
39%
62%
20%
.05
23%
.001

Confidence Gains for Those Whose Confidence Increased, by Specialty

% WHO INCREASED CONFIDENCE

↑ 56% Primary Care Physicians (n= 283)
$P < .001$

↑ 69% Psychiatrists (n= 1,253)
$P < .001$

PRE VS POST CONFIDENCE MEAN & CONFIDENCE GAINS
FOR THOSE WHOSE CONFIDENCE INCREASED

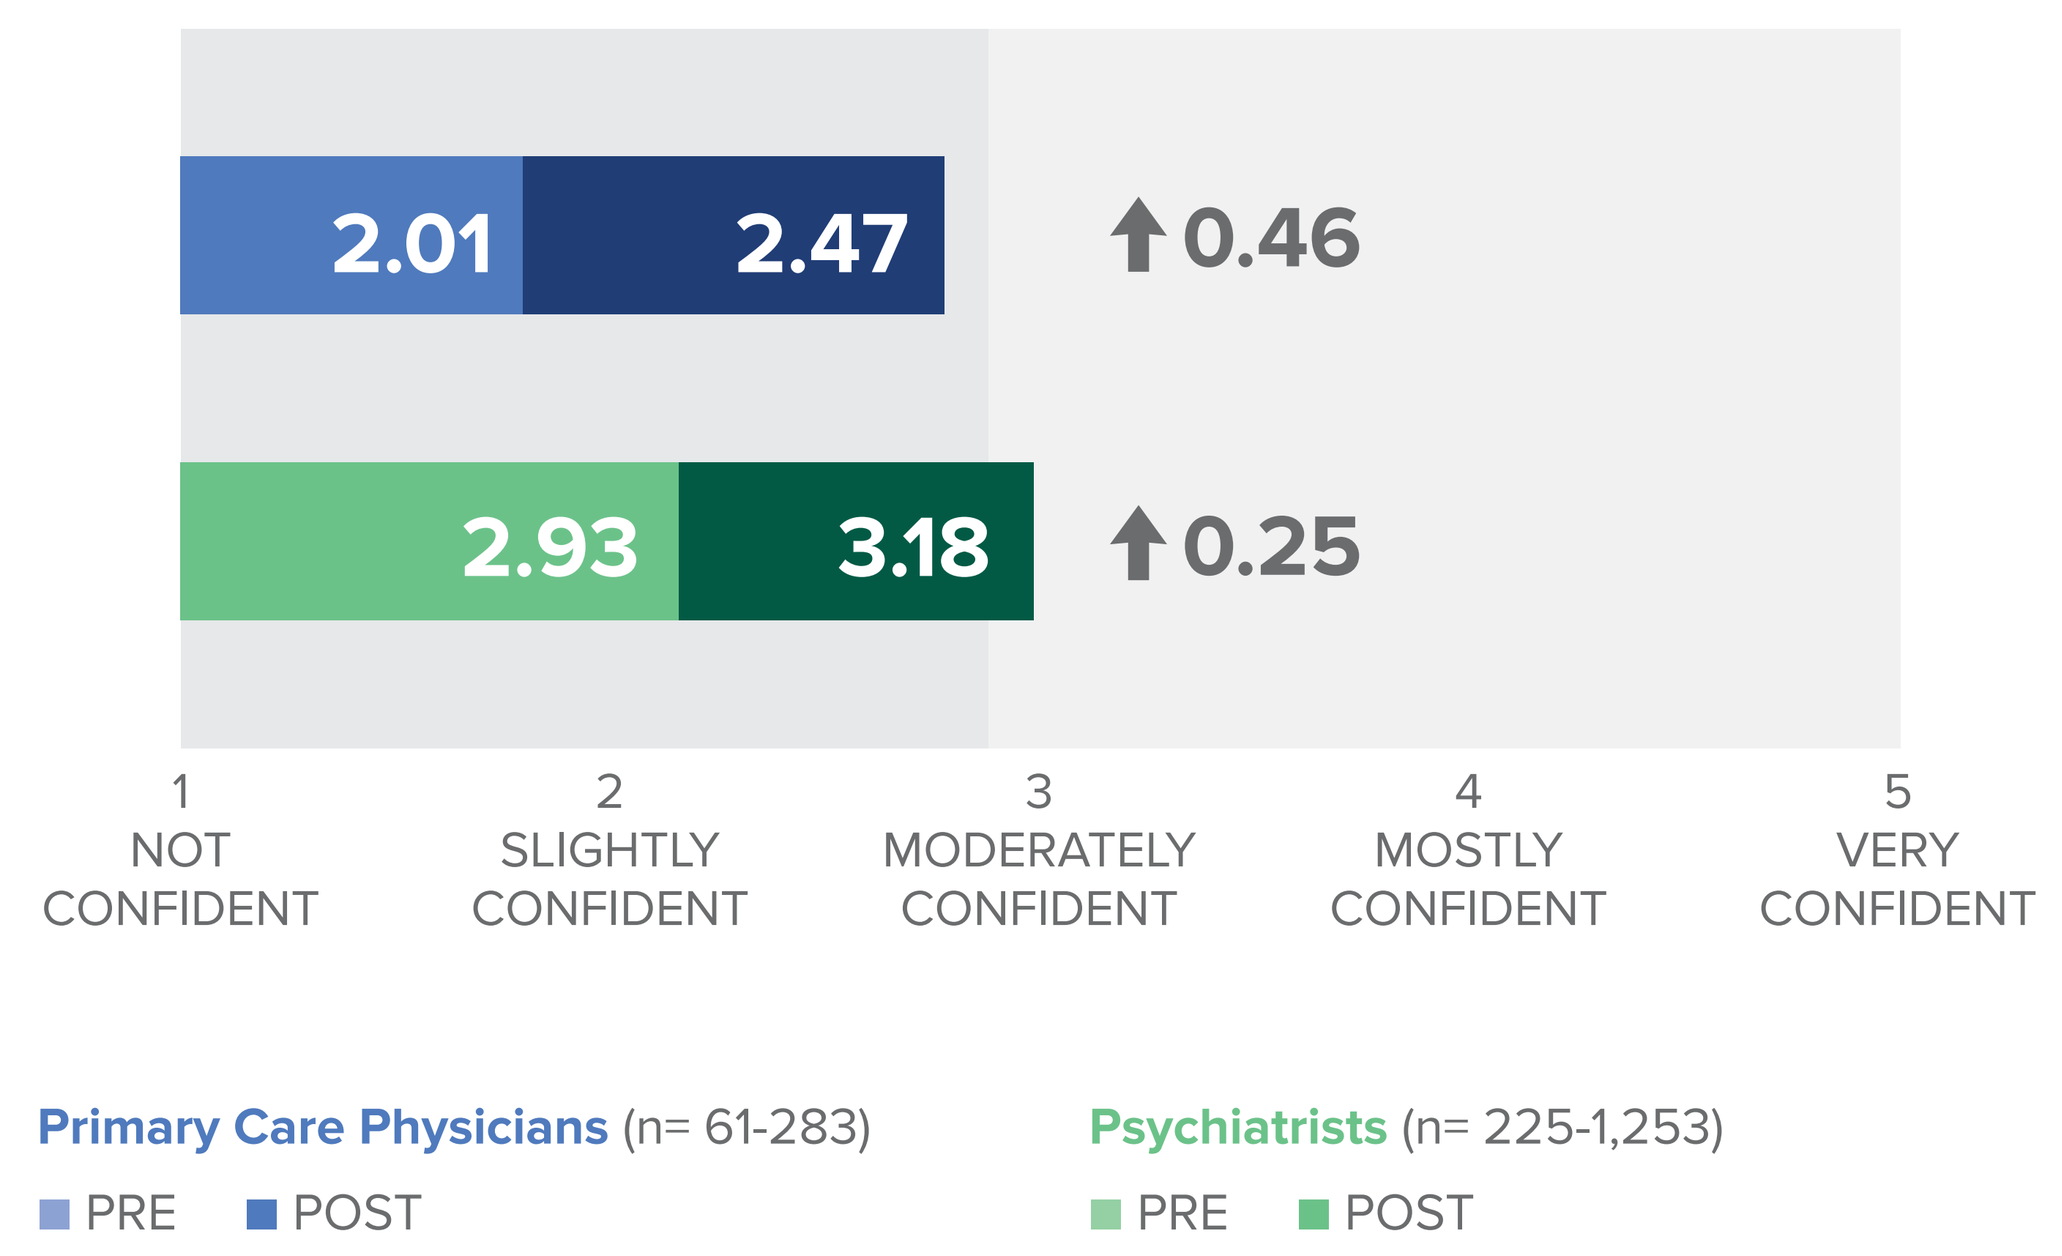
2.01
2.47
0.46
2.93
3.18
0.25
1 NOT CONFIDENT
2 SLIGHTLY CONFIDENT
3 MODERATELY CONFIDENT
4 MOSTLY CONFIDENT
5 VERY CONFIDENT
Primary Care Physicians (n= 61-283)
PRE
POST
Psychiatrists (n= 225-1,253)
PRE
POST

Note: Confidence gains are calculated as absolute change in mean confidence

CONCLUSIONS

Participation in a CME-certified curriculum significantly improved knowledge/competence and confidence among psychiatrist and primary care providers related to individualized care for their patients with schizophrenia. Confidence post-intervention remains only at moderately confident in both learner groups, suggesting that further educational opportunities exist.

ACKNOWLEDGEMENTS

Developed through an independent educational grant from Karuna Pharmaceuticals.

For more information, contact:
Wendy Warfield, MHA, OTR/L, Assoc. Director, Clinical Strategy
wwarfield@medscape.net

Scan here to view this poster online.