Effectiveness of Medical Education at Improving Physician Knowledge, Competence, and Confidence in Identifying Patients Who May Benefit From Novel Therapies for Treatment of Major Depressive Disorder

Medscape EDUCATION

Wendy Warfield, MHA, OTR/L; Sagar V. Parikh, MD, FRCPC; Clinton W. Wright, PharmD, BCPP; Christine Considine: Medscape LLC, Newark, NJ, USA

Presented at **Psych Congress 2024,** October 29-November 1, 2024; Poster #191

BACKGROUND

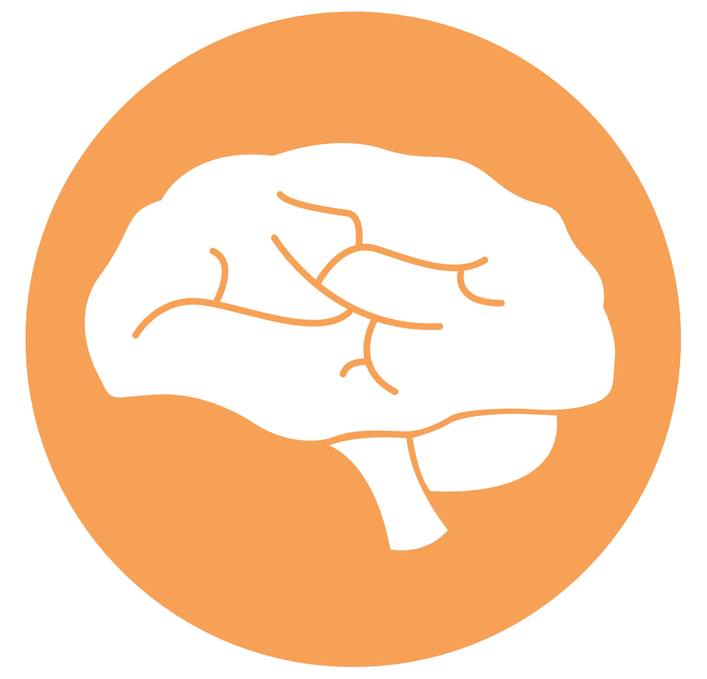

Physicians are challenged to fully understand the pathophysiology of major depressive disorder (MDD) and identify the role and importance of novel therapies. Individualizing treatment for patients living with MDD continues to be a challenge as patients often do not respond to typical first-line treatments that are currently available.

METHODS

This study examined whether online CME could improve the knowledge, confidence, and competence of psychiatrists and primary care physicians in better understanding the pathophysiology of MDD and identifying patients who may benefit from novel therapies.

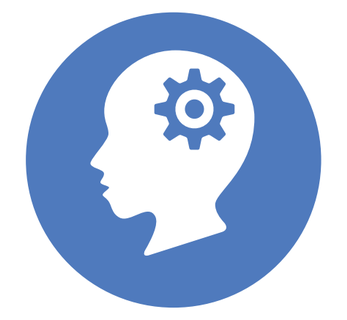
Psychiatrists (n = 1,490)

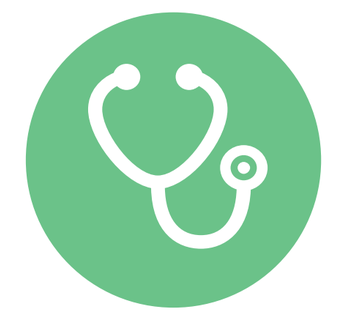
Primary Care Physicians (PCPs) (n = 298)

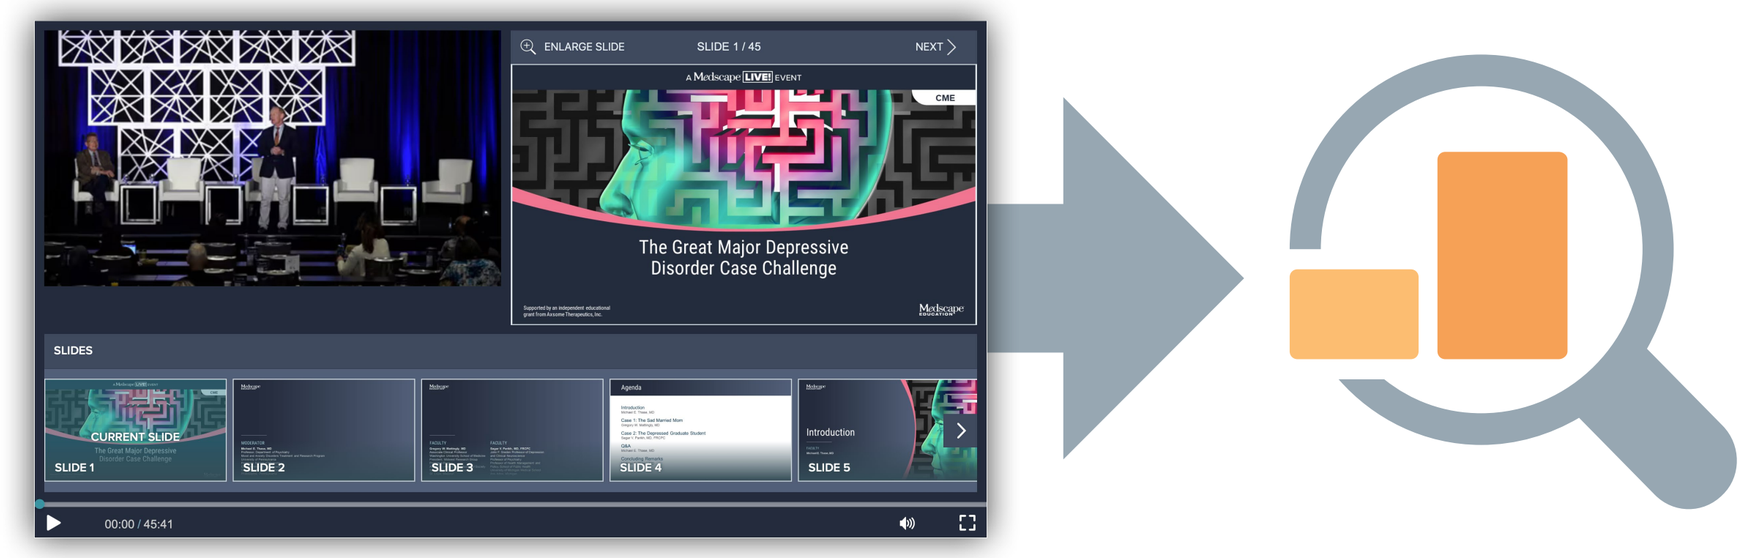
The Great Major Depressive Disorder Case Challenge

How to Read the Linked Learner Assessment

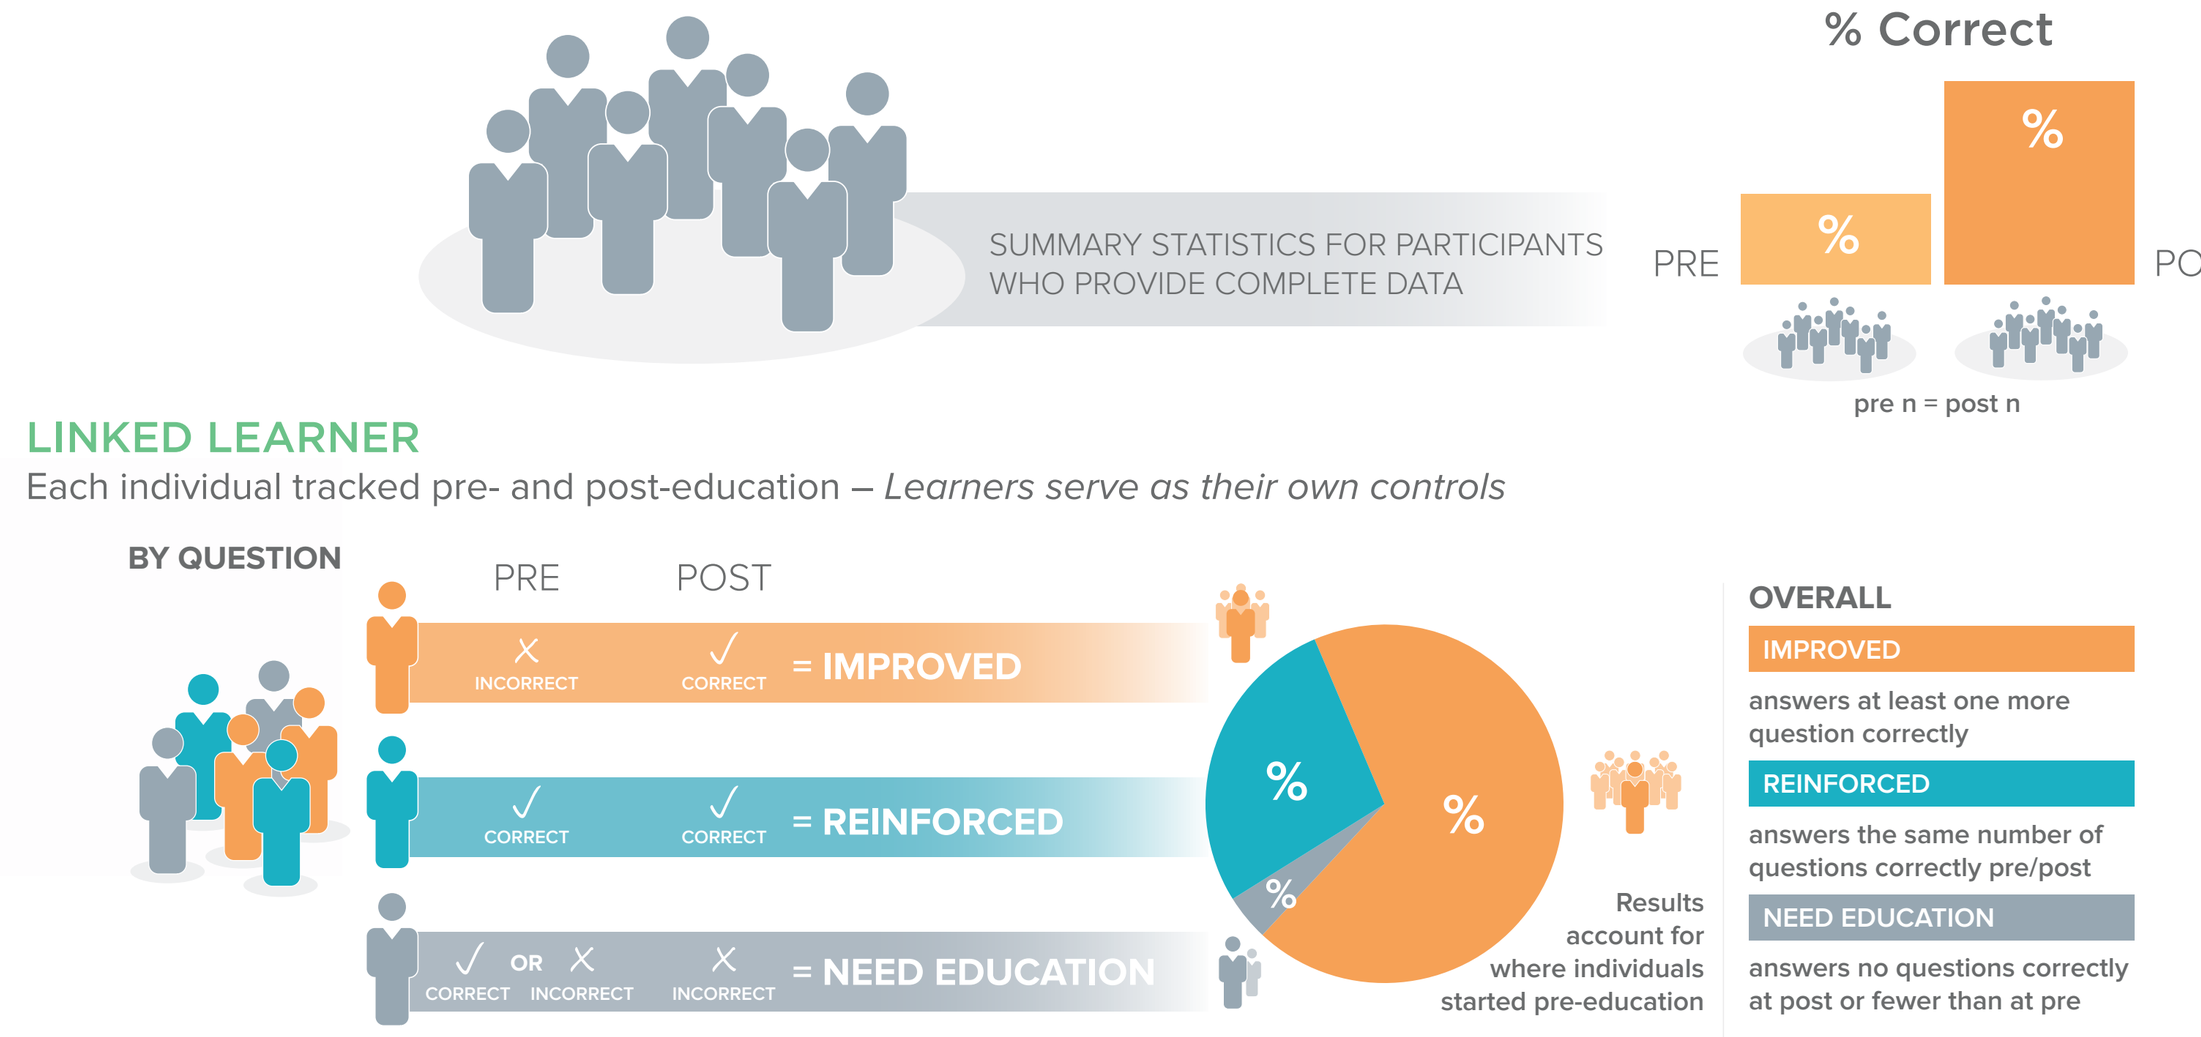
OUTCOMES COMPLETERS
Each individual completed BOTH the pre- and post-education questions – SAME individuals pre- and post-education
% Correct
SUMMARY STATISTICS FOR PARTICIPANTS WHO PROVIDE COMPLETE DATA
PRE
POST
%
%
pre n = post n
LINKED LEARNER
Each individual tracked pre- and post-education – Learners serve as their own controls
BY QUESTION
PRE
POST
INCORRECT
CORRECT
= IMPROVED
CORRECT
CORRECT
= REINFORCED
CORRECT OR INCORRECT
INCORRECT
= NEED EDUCATION
%
%
%
Results account for where individuals started pre-education
OVERALL
IMPROVED
answers at least one more question correctly
REINFORCED
answers the same number of questions correctly pre/post
NEED EDUCATION
answers no questions correctly at post or fewer than at pre

RESULTS

The analysis set consisted of responses from psychiatrist (n = 1490) and primary care physician (n = 298) learner groups who answered all assessment questions during the study period. Analysis of pre- vs post-intervention responses demonstrated a significant improvement in overall knowledge; average correct responses increased from 68% pre-assessment to 78% post-assessment in the psychiatrist learner group and from 53% to 63% in the primary care physician learner group ($P < .001$). Specific areas of improvement include:

- Knowledge regarding the role of glutamatergic dysfunction in MDD pathophysiology: improved from 75% to 80% in the psychiatrist learner group and from 60% to 69% in the primary care provider learner group.
- Greater competence related to identifying patients with MDD who may benefit from a novel antidepressant: improved 53% to 73% in the psychiatrist learner group and from 37% to 52% in the primary care physician learner group.

Post-education, 30% of psychiatrists and 31% of primary care physicians had a measurable increase in their confidence in ability to better understand the pathophysiology of MDD and identify patients who could benefit from a novel antidepressant ($P < .001$).

OVERALL

Psychiatrists (n = 1,490)

AGGREGATED RESULTS

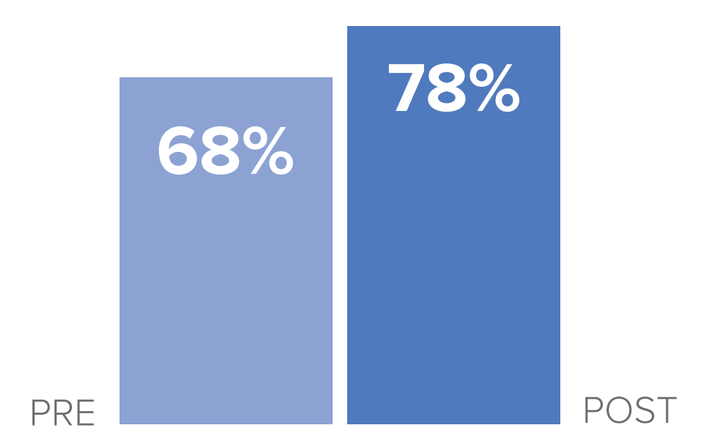
68%
78%
PRE
POST

COHEN'S d

0.53

EFFECT SIZE	EDUCATIONAL IMPACT
< .20	MODEST
.20 - .49	SMALL
.5 - .79	**MODERATE**
≥0.80	LARGE

CHI-SQUARE TEST **$P < .001$**

SIGNIFICANCE (P <.05)

PCPs (n = 298)

AGGREGATED RESULTS

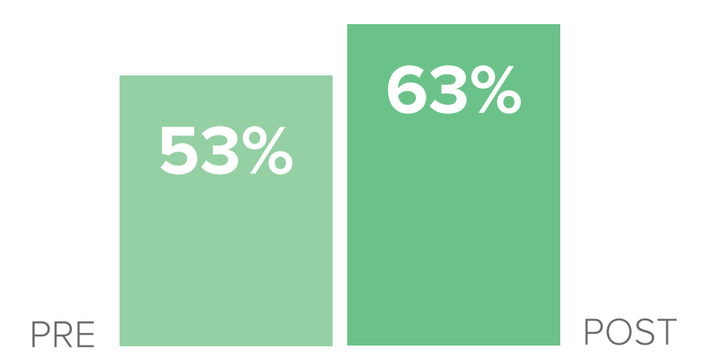
53%
63%
PRE
POST

COHEN'S d

0.49

EFFECT SIZE	EDUCATIONAL IMPACT
< .20	MODEST
.20 - .49	**SMALL**
.5 - .79	MODERATE
≥0.80	LARGE

CHI-SQUARE TEST **$P < .001$**

SIGNIFICANCE (P <.05)

QUESTION 1 RESULTS

QUESTION: Patients with major depressive disorder (MDD) have been found to have: *(Correct Answer: Lower γ-Aminobutyric acid [GABA] levels)*

Psychiatrists (n = 444)

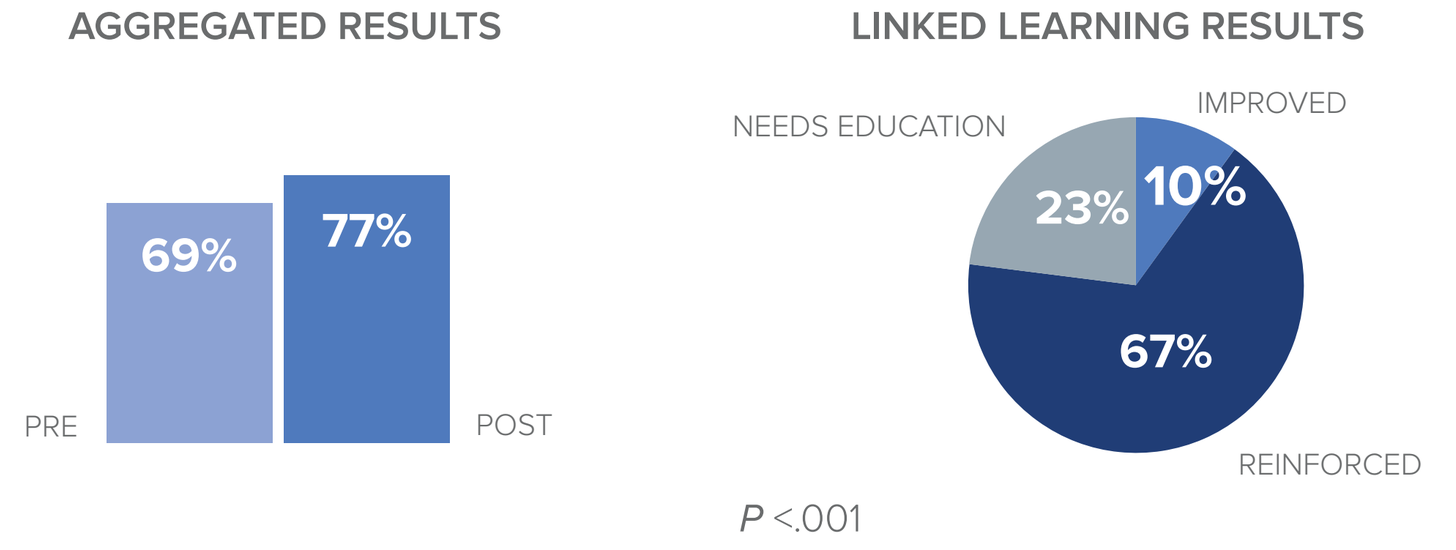
AGGREGATED RESULTS
69%
77%
PRE
POST
LINKED LEARNING RESULTS
NEEDS EDUCATION
IMPROVED
23%
10%
67%
REINFORCED
P <.001

PCPs (n = 86)

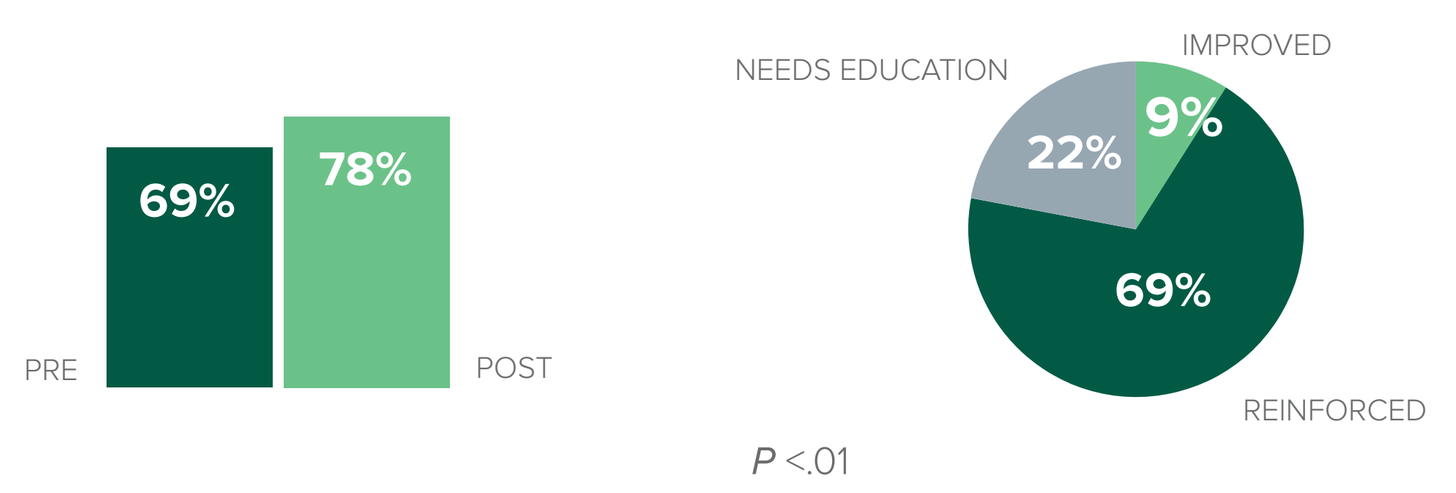
AGGREGATED RESULTS
69%
78%
PRE
POST
LINKED LEARNING RESULTS
NEEDS EDUCATION
IMPROVED
22%
9%
69%
REINFORCED
P <.01

QUESTION 2 RESULTS

QUESTION: A psychiatrist sees a 45-year-old woman for severe MDD. She is currently on venlafaxine 150 mg. She has been experiencing a low mood and lack of interest in anything since she lost her job 7 months ago. She has insomnia, is physically inactive, and reports seeing the future as increasingly bleak. What is the best treatment option for her at this point? *(Correct Answer: Dextromethorphan-bupropion)*

Psychiatrists (n = 444)

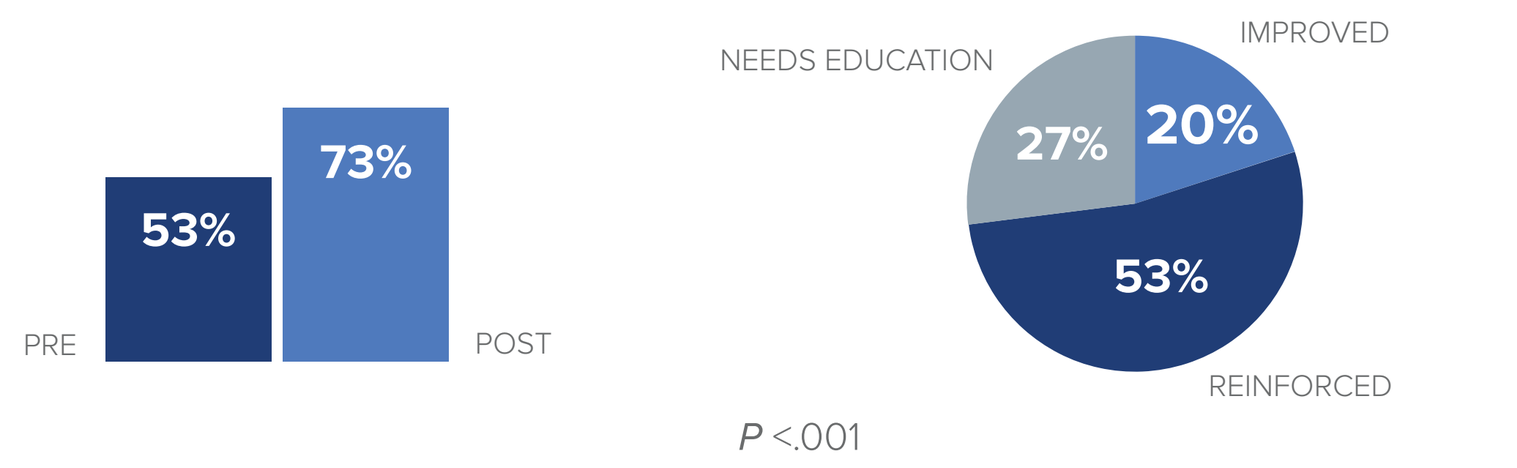
AGGREGATED RESULTS
53%
73%
PRE
POST
LINKED LEARNING RESULTS
NEEDS EDUCATION
IMPROVED
27%
20%
53%
REINFORCED
P <.001

PCPs (n = 86)

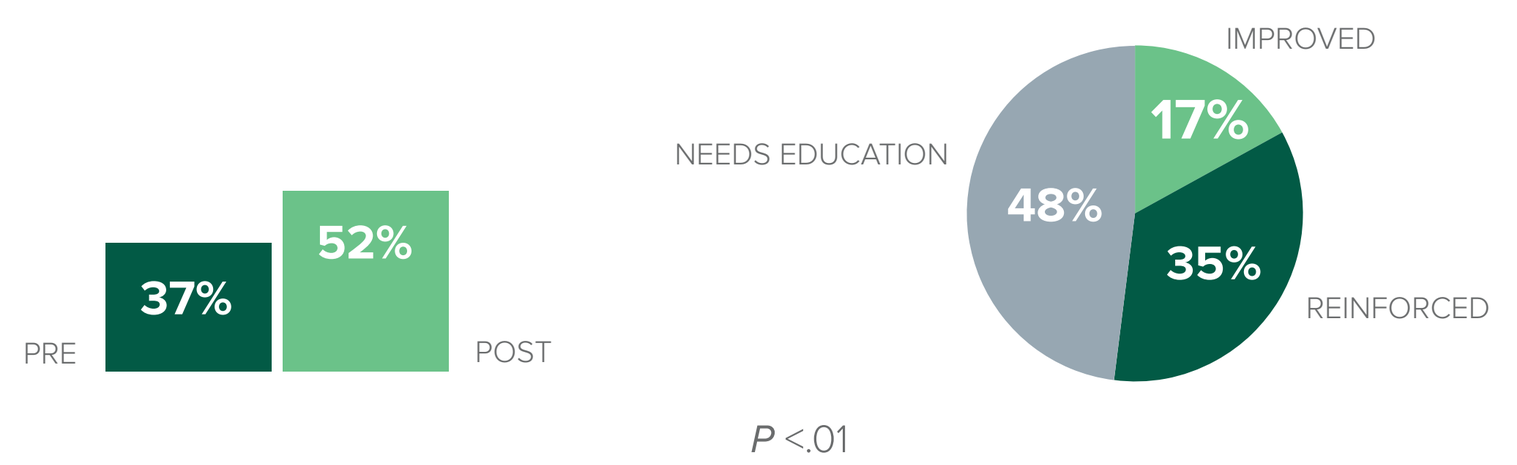
AGGREGATED RESULTS
37%
52%
PRE
POST
LINKED LEARNING RESULTS
NEEDS EDUCATION
IMPROVED
48%
17%
35%
REINFORCED
P <.01

QUESTION 3 RESULTS

QUESTION: Which of the following class of medication is contraindicated with dextromethorphan-bupropion? *(Correct Answer: Monoamine oxidase inhibitors [MAOI])*

Psychiatrists (n = 444)

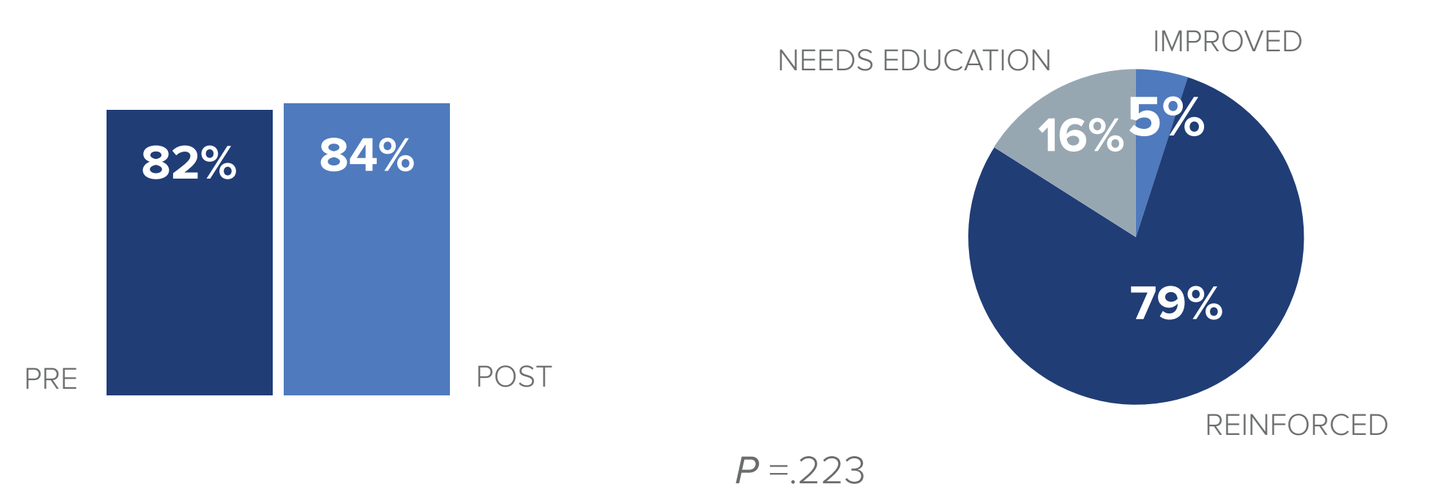
AGGREGATED RESULTS
82%
84%
PRE
POST
LINKED LEARNING RESULTS
NEEDS EDUCATION
IMPROVED
16%
5%
79%
REINFORCED
P =.223

PCPs (n = 86)

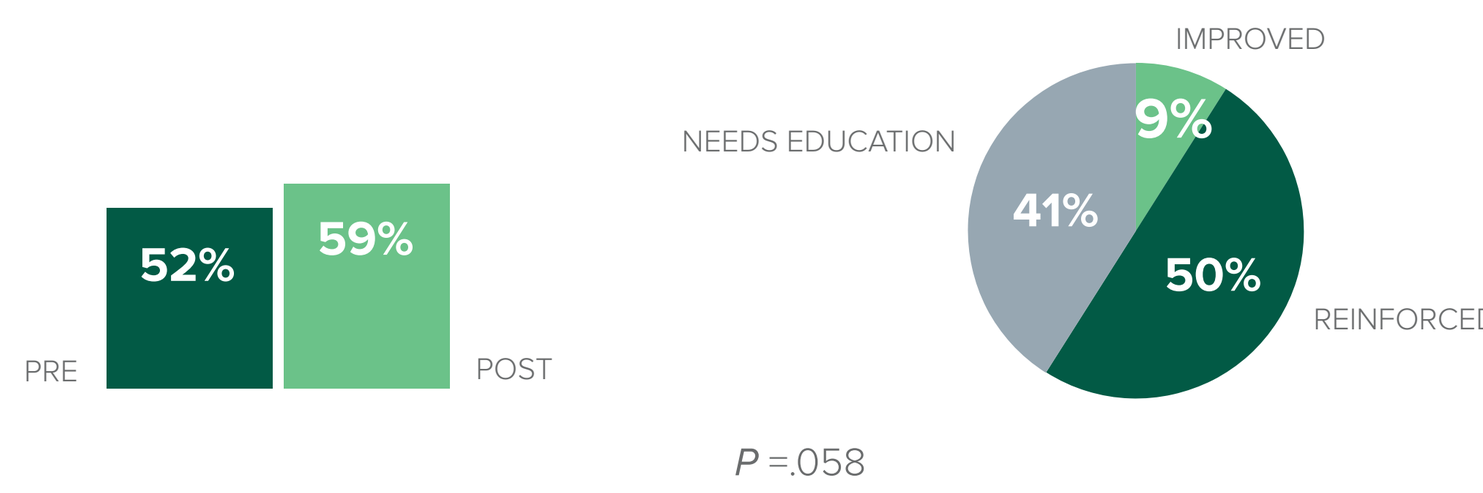
AGGREGATED RESULTS
52%
59%
PRE
POST
LINKED LEARNING RESULTS
NEEDS EDUCATION
IMPROVED
41%
9%
50%
REINFORCED
P =.058

CONFIDENCE ANALYSIS

QUESTION: How confident are you right now in your ability to incorporate antidepressant treatments with novel mechanisms of action into individualized treatment plans for patients with MDD? (Select ranking from 1 [Not confident] to 5 [Very confident])

Psychiatrists (n = 444)

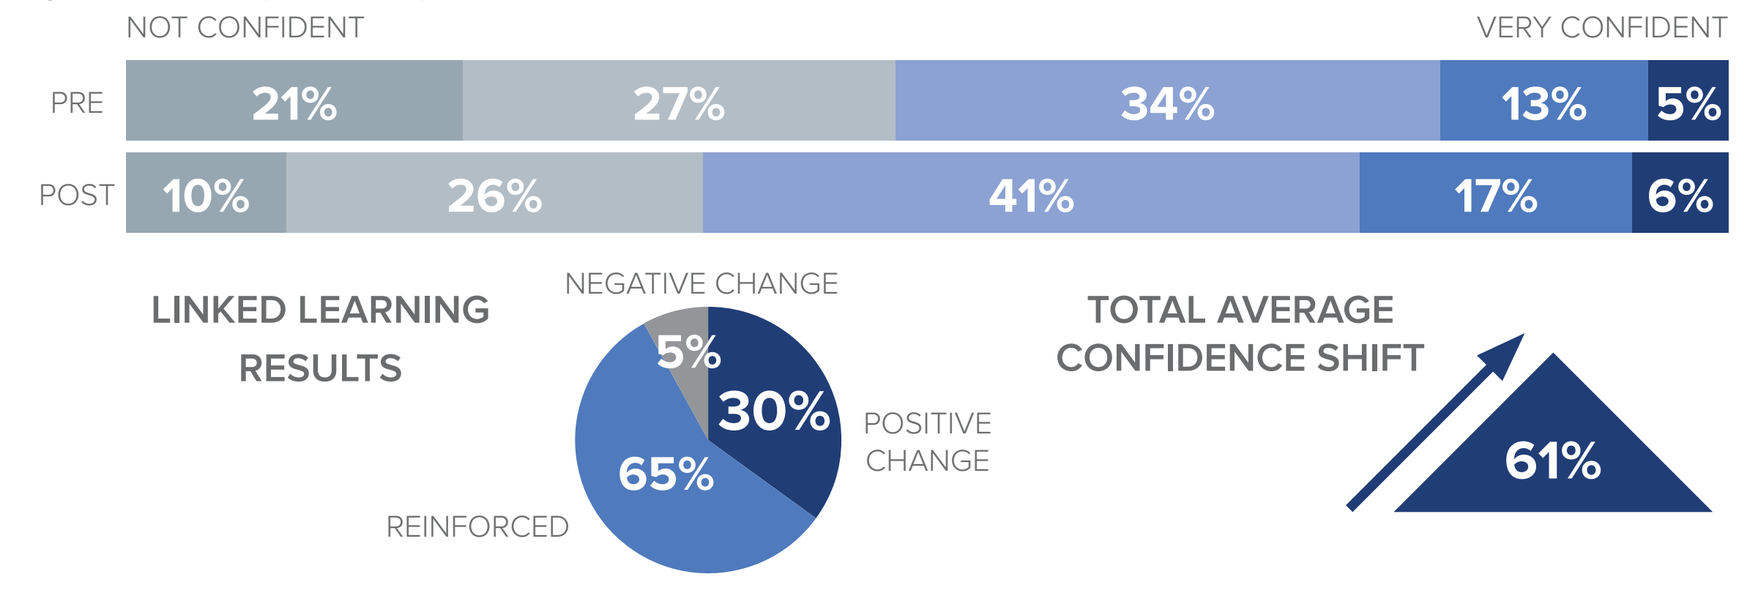
NOT CONFIDENT
VERY CONFIDENT
PRE
21%
27%
34%
13%
5%
POST
10%
26%
41%
17%
6%
LINKED LEARNING RESULTS
NEGATIVE CHANGE
5%
30%
65%
POSITIVE CHANGE
REINFORCED
TOTAL AVERAGE CONFIDENCE SHIFT
61%

PCPs (n = 86)

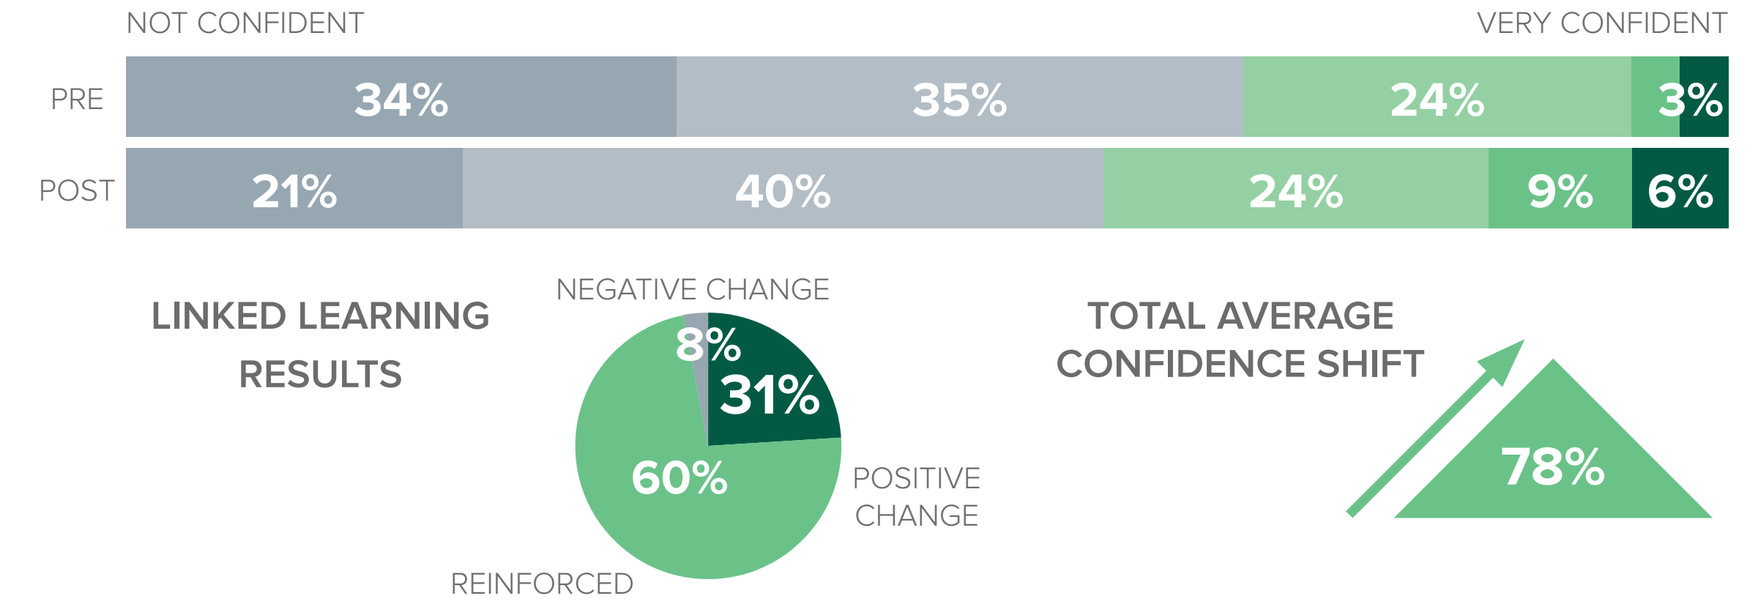
NOT CONFIDENT
VERY CONFIDENT
PRE
34%
35%
24%
3%
POST
21%
40%
24%
9%
6%
LINKED LEARNING RESULTS
NEGATIVE CHANGE
8%
31%
60%
POSITIVE CHANGE
REINFORCED
TOTAL AVERAGE CONFIDENCE SHIFT
78%

CONCLUSIONS

This study demonstrated the success of online CME at improving the knowledge, confidence, and competence of psychiatrists and primary care physicians in their ability to better understand the pathophysiology of MDD and ability to identify how novel therapies may fit into an individualized care plan. Persistent educational gaps were identified for future educational targets.

ACKNOWLEDGEMENTS

Developed through an independent educational grant from Axsome Pharmaceuticals

For more information, contact:

Wendy Warfield, MHA, OTR/L, Assoc. Director, Clinical Strategy

wwarfield@medscape.net

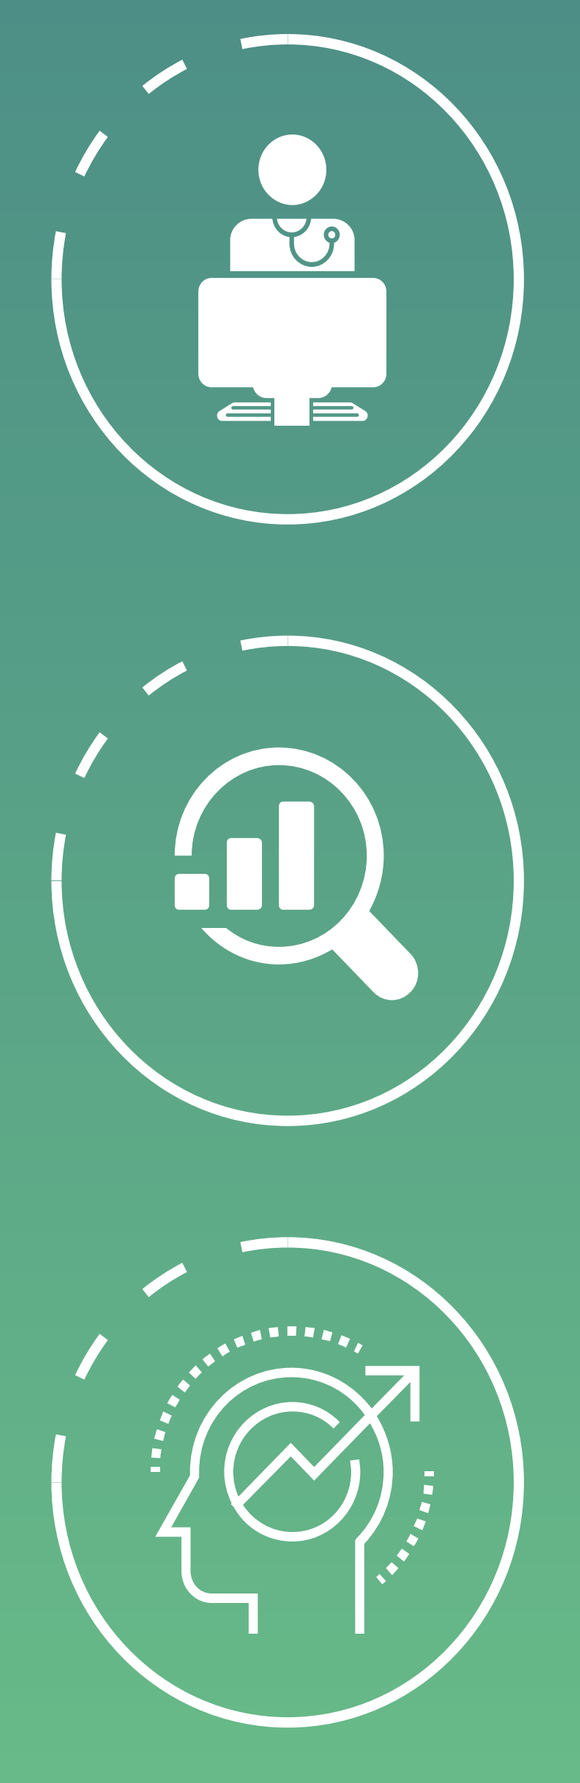

Scan here to view this poster online.