Medscape EDUCATION

Targeted Treatment for Depression: Evaluation of Current Clinical Practices

Jelena Spyropoulos, PhD
Medscape Education, New York, NY

Background

Major depressive disorder (MDD) is the most prevalent mental disorder, affecting approximately 16.2% of adults in the United States.[1] Effective initial and long-term treatment of MDD is associated with several challenges:

- Only 51% of all patients with MDD receive treatment, and only approximately 22% receive adequate care.[1]
- Only half of patients with MDD respond to their first course of therapy, and nearly two-thirds fail to achieve remission with the initial treatment.[2]
- It is often difficult to determine whether a symptom is due to the patient's depression or an adverse effect of the medication.[3]
- Approximately 50% of patients discontinue their antidepressant treatment as a result of tolerability concerns.[3]

Current data ideally allow clinicians to integrate understanding of receptor pharmacology to begin to address specific symptoms or adverse effects; however, studies indicate that few clinicians are sufficiently confident in their knowledge of this issue to fully avail themselves of its practical use; both psychiatrists and primary care physicians (PCPs) have difficulty tailoring therapy based on the pharmacologic profiles of available antidepressant medications.[4]

This study's objective was to increase self-awareness among clinicians of their knowledge, skills, attitudes, practice patterns, and barriers to evaluating, individualizing, and monitoring treatment in patients with MDD.

Methods

A 23-question Clinical Practice Assessment (CPA) survey was developed to assess the current clinical practices and to identify educational needs of psychiatrists and PCPs. The CPA included knowledge- and case-based, multiple-choice questions made available online to healthcare providers without monetary compensation. Confidentiality of CPA respondents was maintained, and responses were de-identified and aggregated before analyses. A coefficient correlation test was performed to determine whether there were relationships between participants' difficulty level related to selection of therapy (CPA question 23) and knowledge of mechanism of action (MOA) (CPA questions 5, 6, 16, 17).

A CME activity evaluation, administered to all participants immediately following CPA completion, determined common barriers to the treatment of MDD, the types of changes participants planned to implement in their practice, and the impact of the CPA content on their practice. A subanalysis was performed to better understand the relationship between CPA responses and CME activity evaluation questions.

Questions in the CPA were scored to derive a combined knowledge score for each participant. This score was used as the dependent variable for subanalysis, using logistic regression in order to determine relationships between knowledge and barriers to effective treatment. A second outcome or dependent variable considered was the number of barriers. This value was treated as a continuous variable in a linear regression analysis used to determine the relationship between the number of barriers and the knowledge score, number of planned changes, and CPA content impact on the practice.

All analyses were two-sided, and coefficients with *P* <.05 were considered significant. A *P* value of .05 < *P* <.10 was considered marginally significant.

The CPA was launched on August 28, 2013 (www.medscape.org/viewarticle/809974), and participant responses were collected over the following 4 weeks.

Results

In total, 273 psychiatrists and 245 PCPs completed the survey.

Figure 1 **Learner specialty.**

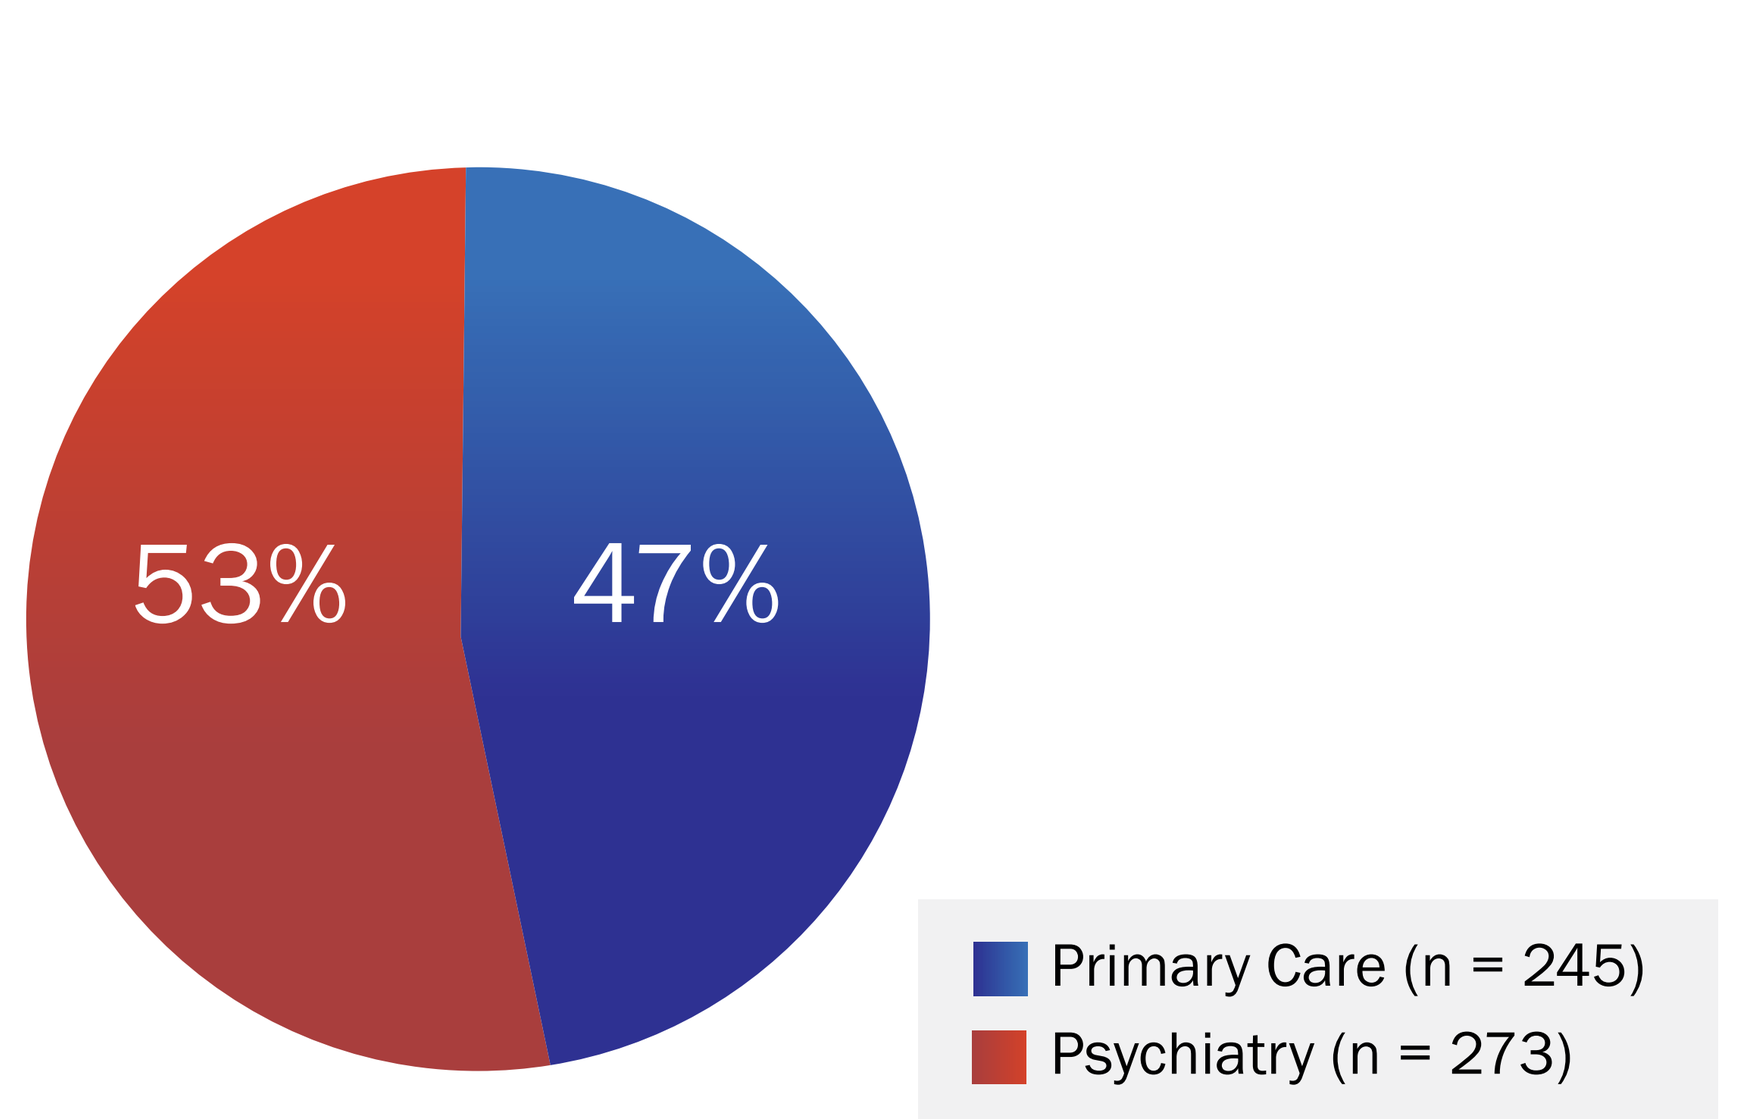

53%
47%
Primary Care (n = 245)
Psychiatry (n = 273)

Cumulative correct scores for all 23 questions averaged 56% among psychiatrists and 45% among PCPs. Clinical assessment scores for each question are represented in Figure 2.

Figure 2 **Clinical assessment scores.**

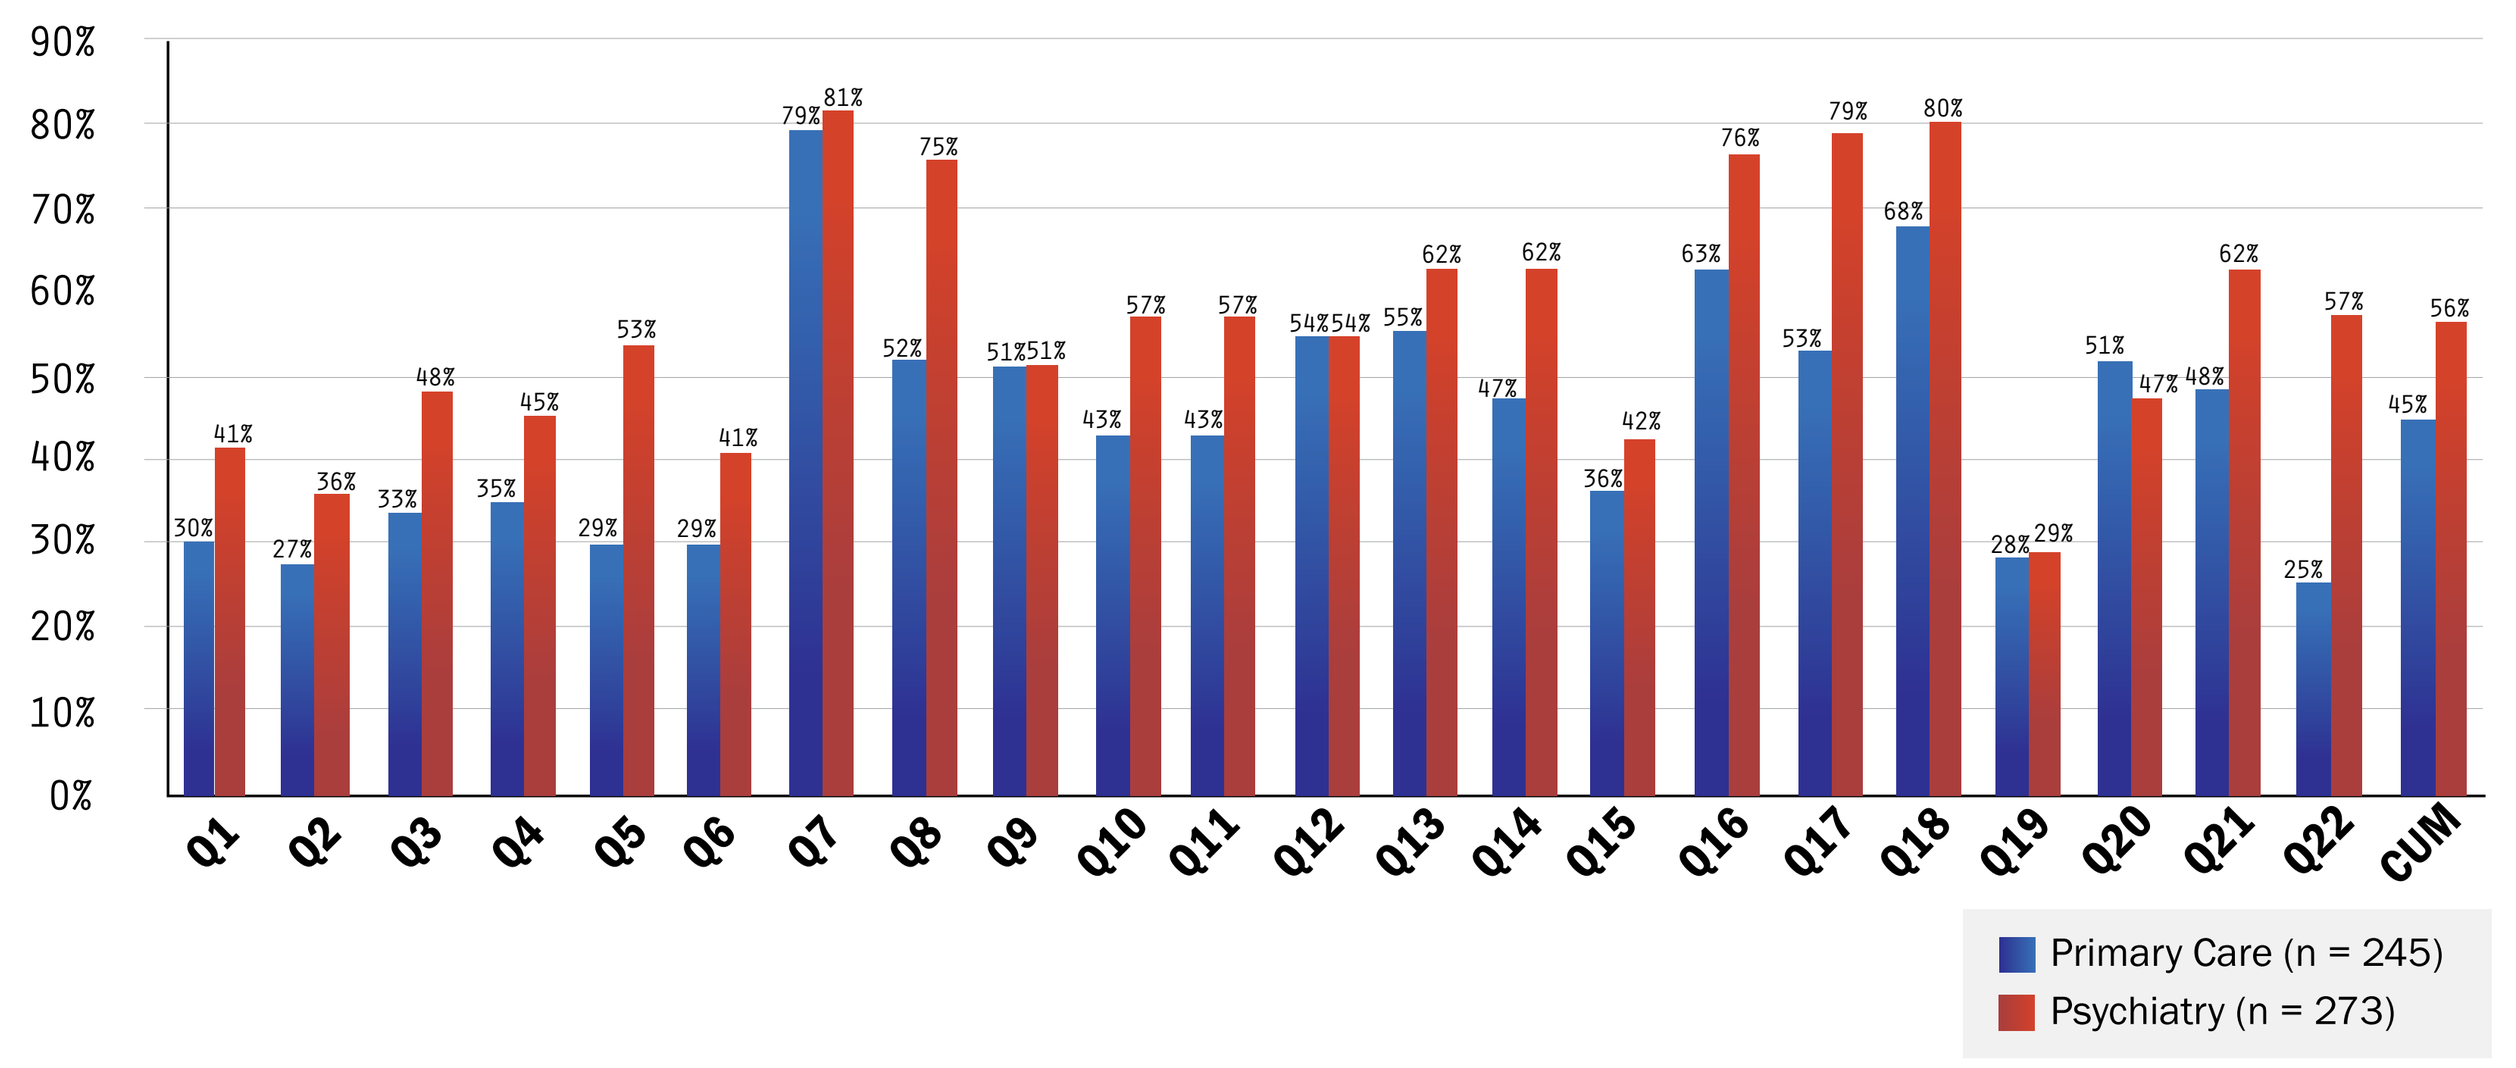

90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
Q1 Q2 Q3 Q4 Q5 Q6 Q7 Q8 Q9 Q10 Q11 Q12 Q13 Q14 Q15 Q16 Q17 Q18 Q19 Q20 Q21 Q22 CUM
30% 41%
27% 36%
33% 48%
35% 45%
29% 53%
29% 41%
79% 81%
52% 75%
51% 51%
43% 57%
43% 57%
54% 54%
55% 62%
47% 62%
36% 42%
63% 76%
53% 79%
68% 80%
28% 29%
51% 47%
48% 62%
25% 57%
45% 56%
Primary Care (n = 245)
Psychiatry (n = 273)

The assessment questions were further categorized according to the activity learning objectives. The learning objectives and the questions related to them are:

1. Interpret data on biochemical and neurotransmitter function in depression and the relation to current theories of treatment; Questions 1, 2, 3, 4, 5, 6, 7
2. Formulate treatment decisions based on use of scales to assess depression symptoms; Questions 8, 9, 10, 11, 12, 13
3. Individualize initial and ongoing strategies for the treatment of patients with depression; Questions 14, 15, 16, 17, 18, 19, 20, 21, 22

In general, PCPs scored highest on LO 2 related to the use of assessment tools. Psychiatrists scored higher on questions related to LO 3 focused on individualizing treatment compared with questions related to the other 2 LOs.

Figure 3 **Average assessment scores by learning objectives.**

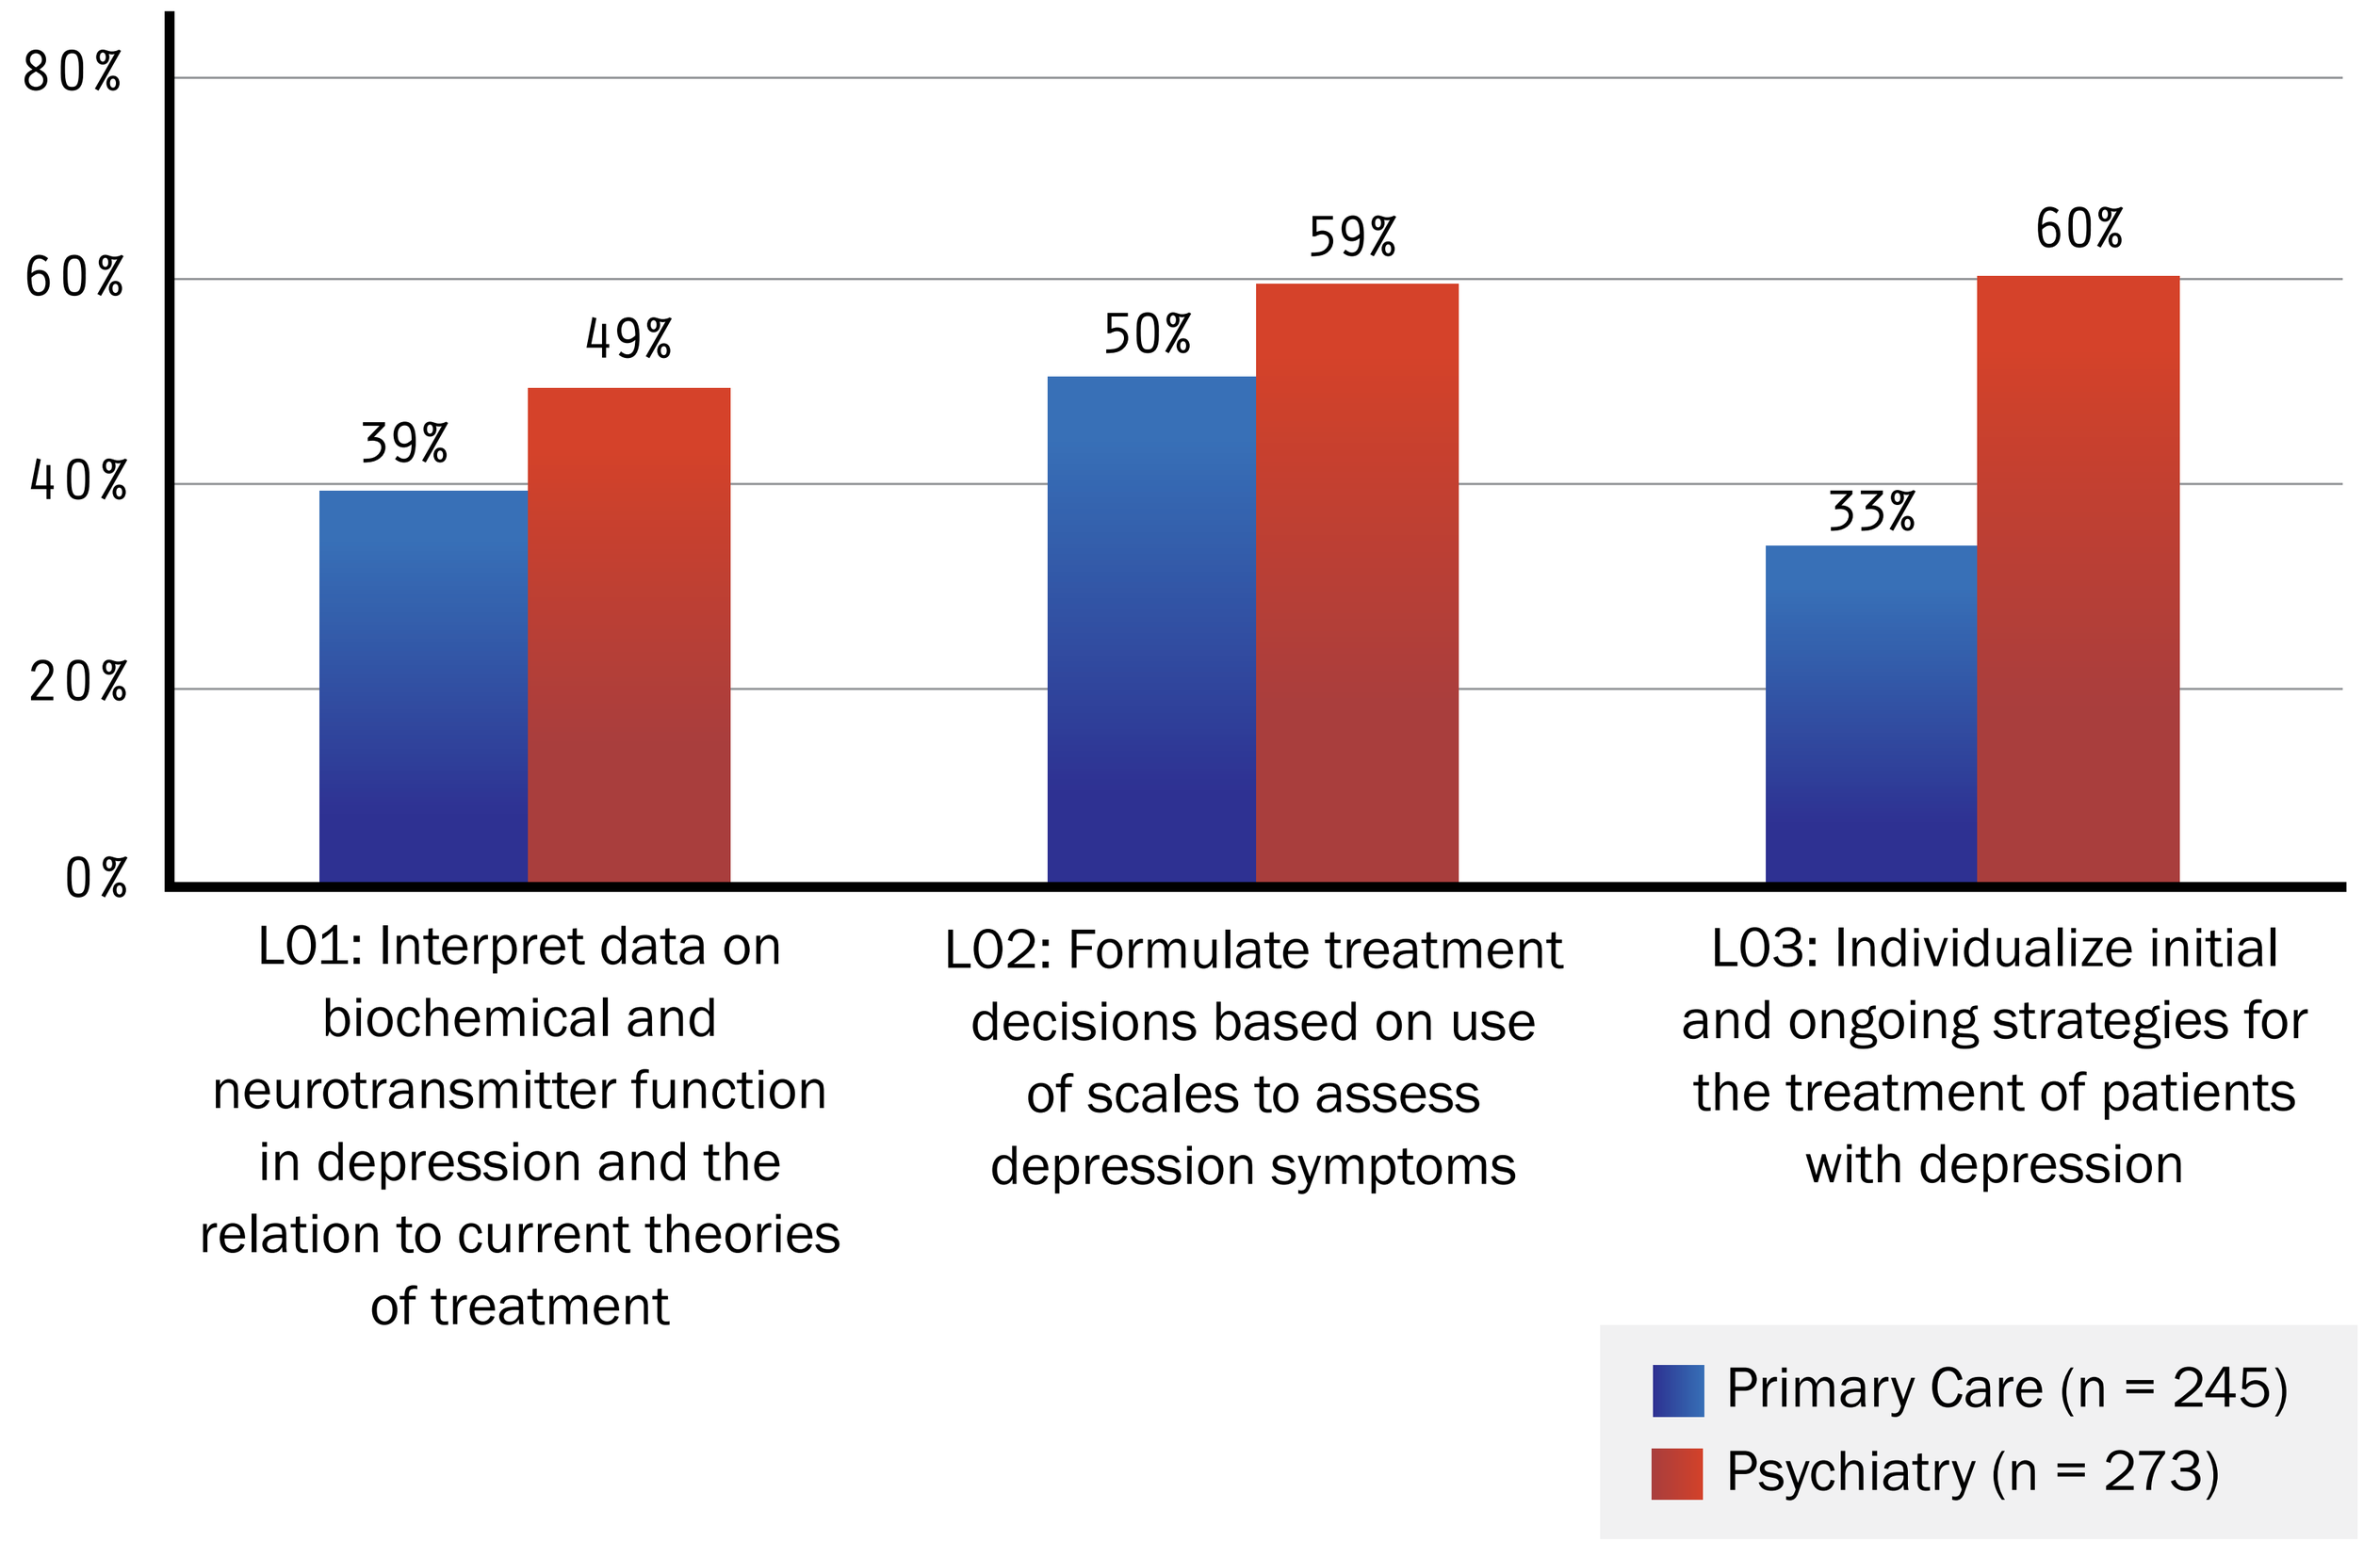

80%
60%
40%
20%
0%
39%
49%
50%
59%
33%
60%
LO1: Interpret data on biochemical and neurotransmitter function in depression and the relation to current theories of treatment
LO2: Formulate treatment decisions based on use of scales to assess depression symptoms
LO3: Individualize initial and ongoing strategies for the treatment of patients with depression
Primary Care (n = 245)
Psychiatry (n = 273)

Educational Gaps:

Based on the assessment questions, educational gaps were uncovered in both psychiatry and PCP groups (Table 1).

Table 1 **Average Correct Scores by Educational Gap**

Educational Gap	Average Correct Scores (Psychiatrists)	Average Correct Scores (PCPs)
Understanding of the theoretical basis of individual patients' MDD and integrating this knowledge into treatment selection	44%	30%
Appropriate use of patient-reported depression rating scales to discern disability or quality of life issues	42%	48%
Use of appropriate therapy in patients with comorbidities	44%	43%
Management of treatment-related depression	49%	33%

Inverse Correlation Between Knowledge of MOA and Difficulty in Selection of Therapy

Respondents were asked to rank the level of difficulty (with 1 being the easiest and 7 being the most difficult) they believe they encounter in selecting first-line antidepressant therapy in patients with MDD, based on the MOA of currently available agents. Although the highest percentage in both specialties indicated difficulty on a level of 4 out of 7 (29% of PCPs, 25% of psychiatrists), PCPs reported greater difficulty overall than psychiatrists (Figure 3).

When a correlation coefficient test was run on the responses to question 23 and the responses to questions related to MOA (questions 5, 6, 16, 17), results showed that all participants, including both specialty groups who reported less difficulty in selecting therapy (question 23), had significantly higher knowledge of MOA (more correct answers on MOA questions: 5, 6, 16, 17, *P* <.05).

Figure 4 **Difficulty level.**

Q23 How difficult is the selection of first-line antidepressant therapy in patients with MDD based on the mechanisms of action of currently available agents?

(Select ranking from 1 [Easy] to 7 [Difficult])

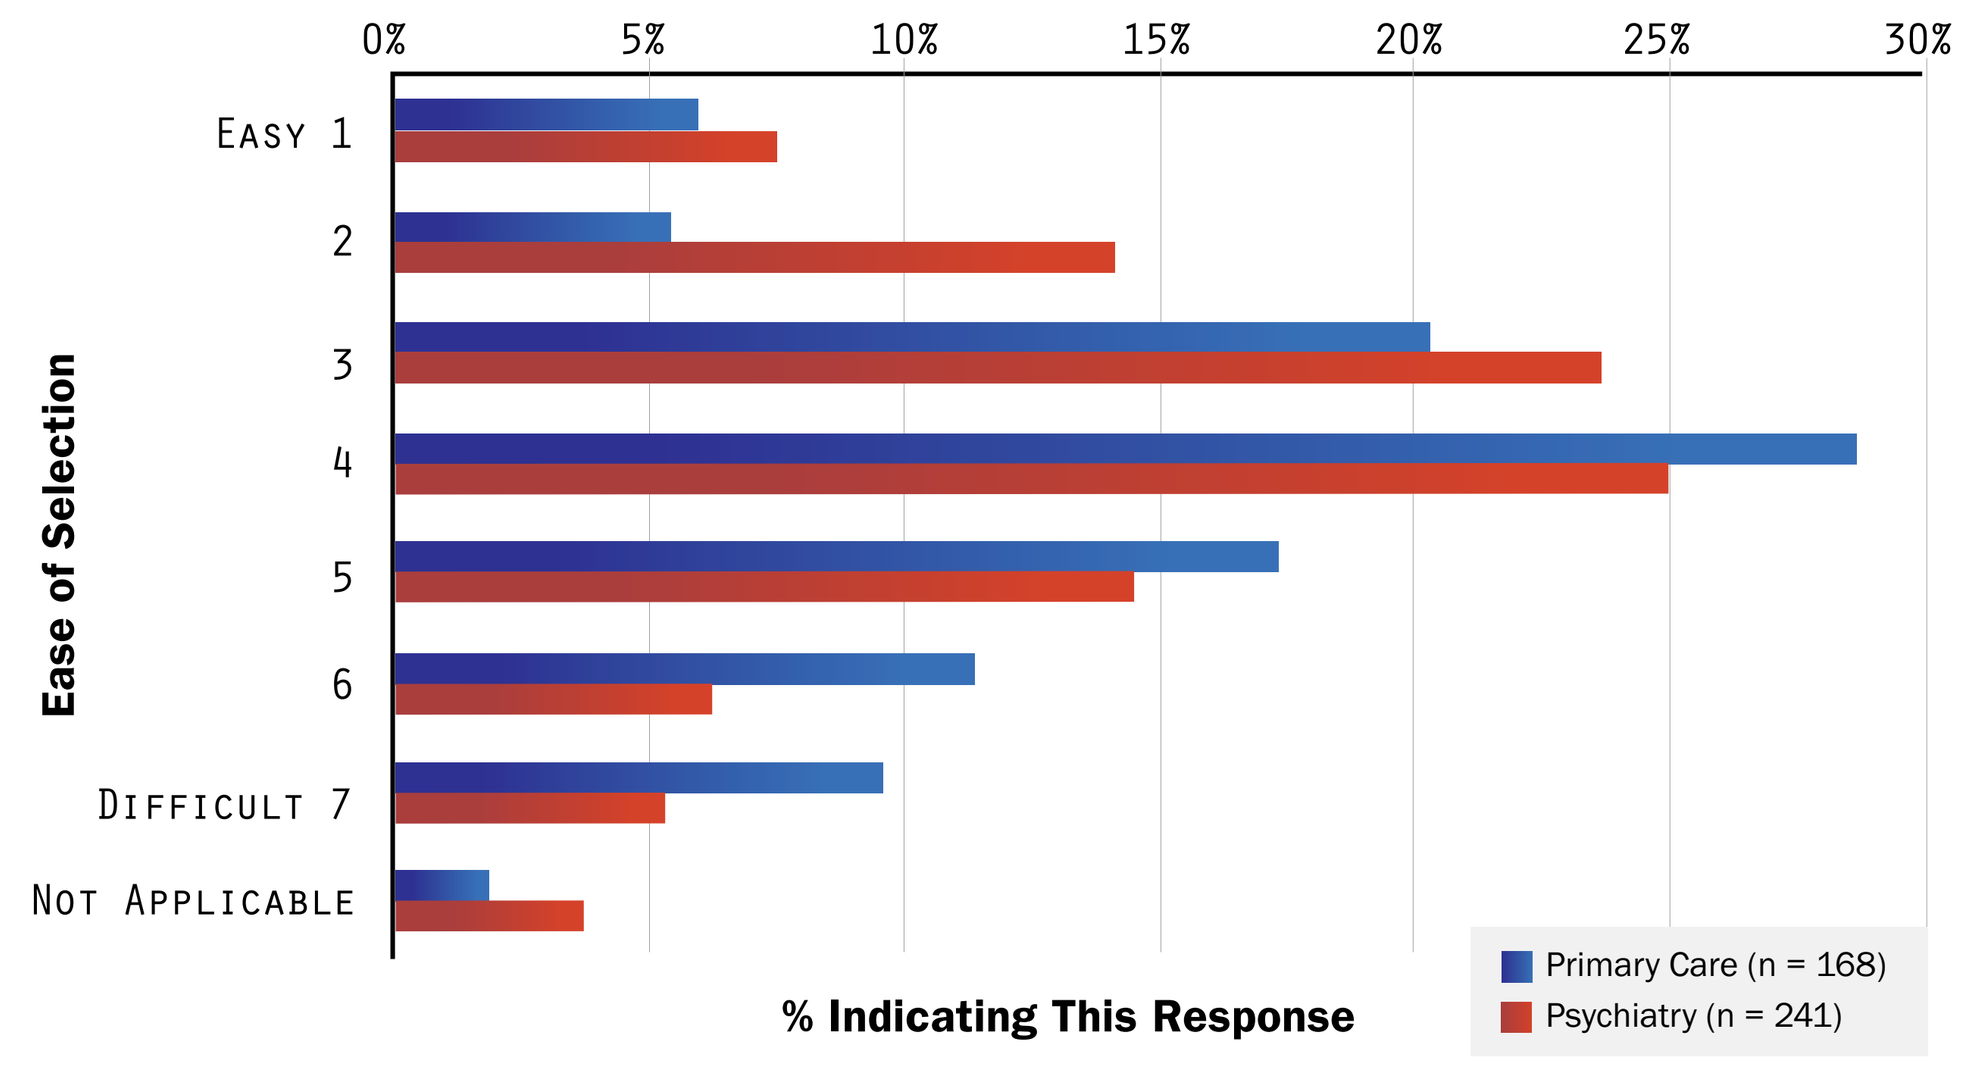

0%
5%
10%
15%
20%
25%
30%
Easy 1
2
3
4
5
6
Difficult 7
Not Applicable
Ease of Selection
% Indicating This Response
Primary Care (n = 168)
Psychiatry (n = 241)

Positive Correlation Between Knowledge and Impact on Practice

Respondents in both specialty groups who agreed or strongly agreed that the CPA content had an impact on their practice had a higher overall knowledge score in the CPA that those who disagreed or strongly disagreed that the CPA content had an impact on their practice.

Positive Correlation Between Planned Changes and Number of Barriers

In the second analysis, participants from both specialty groups who indicated that they would implement a higher number of planned changes in their clinical practice also anticipated a higher number of barriers to implement those changes, indicating a positive correlation between planned changes and number of barriers (*P* <.05).

Conclusions

Medscape's large membership of psychiatrists and PCPs allowed for a robust analysis of the data. The instructional design of the CPA allowed for both the identification of educational gaps related to MDD management as well as for the reinforcement of key educational concepts tailored to elicit appropriate HCP practice changes. Results from this assessment were consistent with knowledge gaps identified in the Medscape Education needs assessment for this program. Baseline knowledge scores in the management of depression were low for psychiatrists as well as PCPs.

Results of statistical subanalyses showed that:

- The cumulative knowledge score was positively correlated to the question regarding the impact of the CPA on the readers' practice. Thus, those who scored higher on the assessment feel more strongly that the content will impact their practice.
- The number of planned changes in practice corresponded to a higher number of barriers, indicating that those who are planning a greater number of changes also anticipate encountering more barriers.
- Physicians who answered more incorrect questions related to MOA reported greater difficulty selecting first line-therapy based on MOA.

This assessment of healthcare providers' clinical practices identified knowledge gaps that support the need to develop educational interventions on MDD management, including:

- Understanding theoretical causes of depressive symptoms and selecting treatment
- Best practices for incorporating rating scales into clinical practice
- Treatment selection taking into account MOAs of available therapies
- Treatment selection taking into account comorbidities and treatment-resistant depression

References

1. Kessler RC, Berglund P, Demler O, et al; National Comorbidity Survey Replication. The epidemiology of major depressive disorder: results from the National Comorbidity Survey Replication (NCS-R). *JAMA*. 2003;289:3095-3105.
2. Rush AJ, Trivedi MH, Wisniewski SR, et al. Acute and longer-term outcomes in depressed outpatients requiring one or several treatment steps: a STAR*D report. *Am J Psychiatry*. 2006;163:1905-1917.
3. Connolly KR, Thase ME. Emerging drugs for major depressive disorder. *Expert Opin Emerg Drugs*. 2012;17:105-126.
4. Medscape Education Survey. Snapshot of depression survey 2012 [.pdf file]. SurveyMonkey.com. October 2012. Accessed February 12, 2014.

Notes

For more information, contact Jelena Spyropoulos, PhD, Director of Clinical Strategy, Medscape, LLC at jspyropoulos@medscape.net.

Source of Support

This CME-certified activity was supported by an independent educational grant from Takeda Pharmaceuticals North America and Lundbeck.

Scan here to view this poster online.